ID # 862

Peripheral Blood Mononuclear Cells and Plasmacytoid Dendritic Cells from Healthy Human Females Exhibit Altered TLR7-Mediated Immune Responses Compared to Males

Melissa Precopio, Dong Yu, Ekambar R. Kandimalla, and Sudhir Agrawal

Idera Pharmaceuticals, Inc., 167 Sidney Street, Cambridge, MA 02139

INTRODUCTION

Toll-like receptors (TLRs) are a family of pattern recognition receptors expressed by cells of the immune system (1). TLR7 and 9 are expressed in endosomal membranes of human plasmacytoid dendritic cells (pDCs) and B cells and recognize nucleic acid molecular patterns (2-4). TLR7 is a receptor for viral single-stranded RNA, while TLR9 is a receptor for bacterial DNA containing unmethylated CpG motifs (5,6).

Recognition of nucleic acids by TLR7 and 9 is important in mediating appropriate immune responses against bacterial and viral infections. Endosomal location of these TLRs makes these receptors inaccessible to self-DNA and -RNA. However, under certain pathologic conditions such as systemic lupus erythematosus (SLE), circulating immune complexes containing self-DNA or -RNA can induce proinflammatory cytokines through TLR7 and 9 (7,8). Interestingly, most autoimmune diseases, including SLE, disproportionally affect females.

We have undertaken this study to understand if there are differences in TLR7- and TLR9-mediated immune responses between males and females. We measured immune responses induced *in vitro* by a TLR7 agonist (9) and a TLR9 agonist (10). Cytokine induction and costimulatory marker upregulation was assessed in response to each TLR agonist in PBMCs and pDCs of healthy males and females. Additionally, cytokine induction in response to TLR7 or 9 agonist treatment of PBMCs was also compared between healthy and SLE-afflicted females.

TLR7 and TLR9 as Receptors of Endogenous Danger-Associated Molecular Patterns (DAMPs)

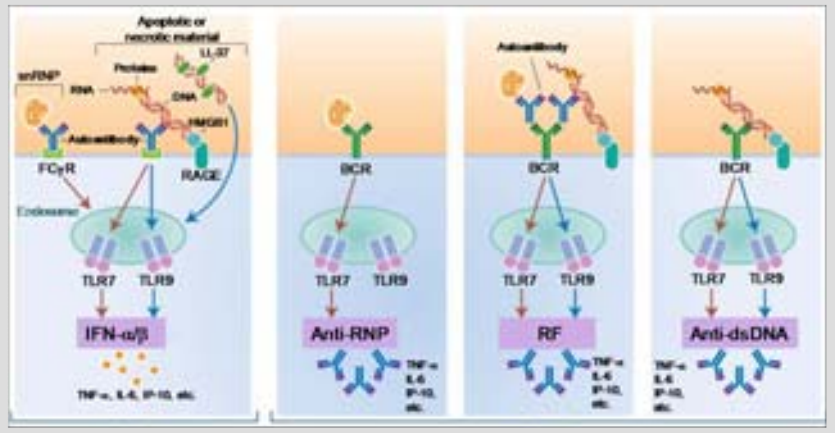
TLR7
TLR9
IFN-α/β
Anti-RNP
RF
Anti-dsDNA
BCR
RAGE

pDC B cell

RESULTS

Agonist-Induced Cytokine Dose-Response Curves in PBMCs

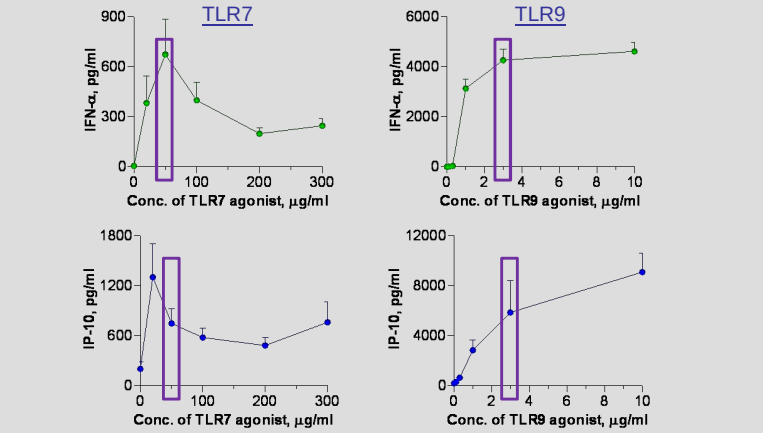
TLR7
TLR9
IFN-α, pg/ml
IP-10, pg/ml
Conc. of TLR7 agonist, μg/ml
Conc. of TLR9 agonist, μg/ml

Based on the dose-response curves, we chose 50 μg/ml TLR7 agonist and 3 μg/ml TLR9 agonist for further studies in PBMCs

TLR7 Agonist-Induced Cytokines in pDCs from Healthy Females and Males

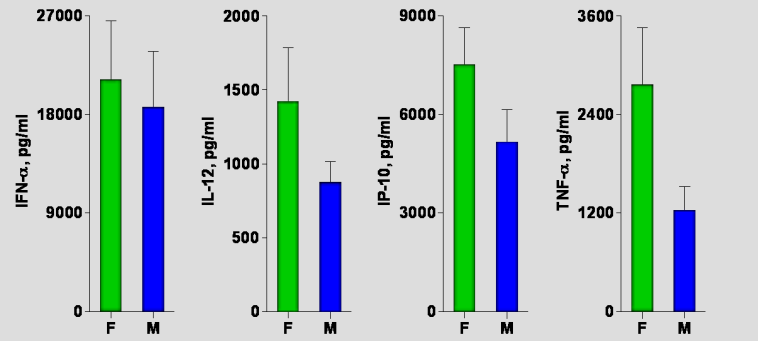
IFN-α, pg/ml
IL-12, pg/ml
IP-10, pg/ml
TNF-α, pg/ml
F
M

Incubation of pDCs with TLR7 agonist results in higher IFN-α, IL-12, IP-10, and TNF-α secretion in females compared to males

TLR7 and TLR9 Agonist-Induced Activation Marker Expression in pDCs from Healthy Females and Males

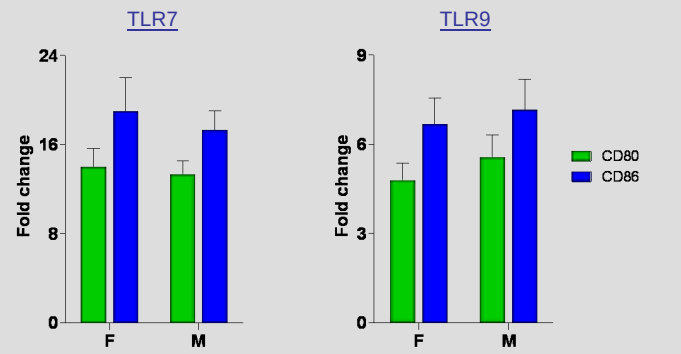
TLR7
TLR9
Fold change
CD80
CD86
F
M

Incubation of pDCs with TLR7 agonist or TLR9 agonist results in increased expression of CD80 and CD86 to similar levels in females and males

TLR7 Agonist-Induced Cytokines in PBMCs from Healthy Females and Males

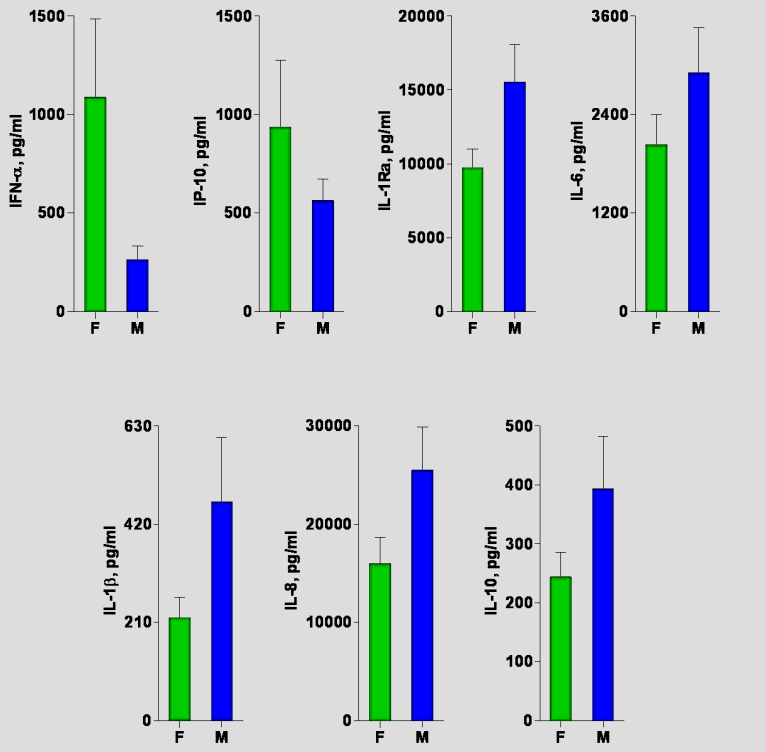
IFN-α, pg/ml
IP-10, pg/ml
IL-1Ra, pg/ml
IL-6, pg/ml
IL-1β, pg/ml
IL-8, pg/ml
IL-10, pg/ml
F
M

Incubation of PBMCs with TLR7 agonist results in higher IFN-α and IP-10 secretion and lower IL-1Ra, IL-6, IL-1β, IL-8, and IL-10 secretion in females compared to males

TLR9 Agonist-Induced Cytokines in PBMCs and pDCs from Healthy Females and Males

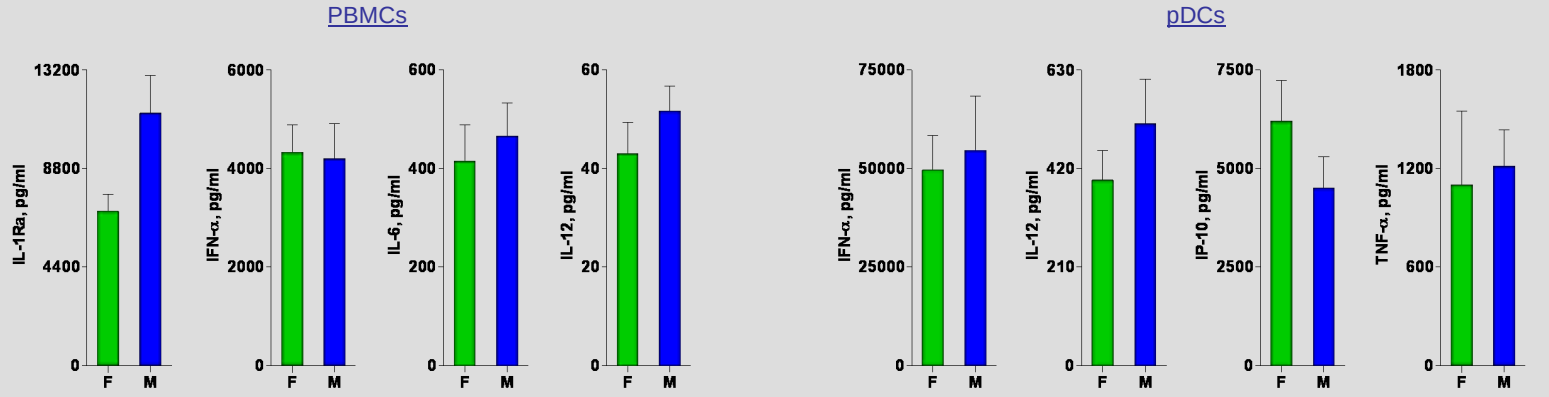
PBMCs
pDCs
IL-1Ra, pg/ml
IFN-α, pg/ml
IL-6, pg/ml
IL-12, pg/ml
IP-10, pg/ml
TNF-α, pg/ml
F
M

Incubation of PBMCs with TLR9 agonist results in similar levels of cytokine secretion in females compared to males, with the exception of higher IL-1Ra levels in males

Incubation of pDCs with TLR9 agonist results in similar levels of cytokine secretion in females compared to males

TLR7 Agonist-Induced Cytokines in PBMCs from Healthy and SLE-Afflicted Females

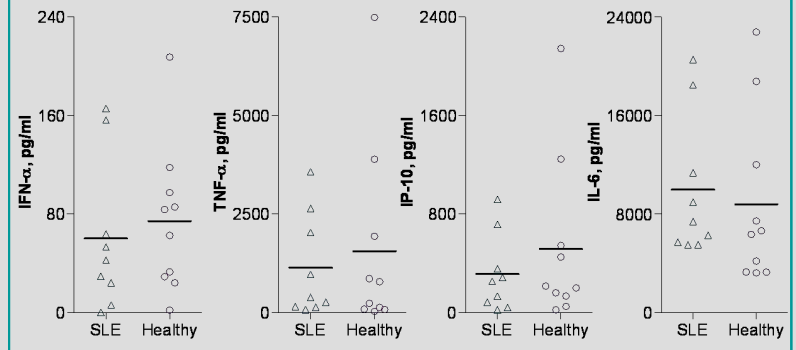
IFN-α, pg/ml
TNF-α, pg/ml
IP-10, pg/ml
IL-6, pg/ml
SLE
Healthy

Incubation of PBMCs with TLR7 agonist results in lower levels of IFN-α, TNF-α, and IP-10 secretion in SLE patients compared with healthy controls

TLR9 Agonist-Induced Cytokines in PBMCs from Healthy and SLE-Afflicted Females

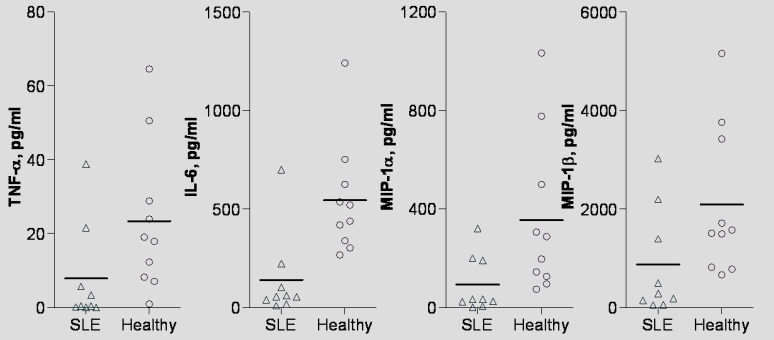
TNF-α, pg/ml
IL-6, pg/ml
MIP-1α, pg/ml
MIP-1β, pg/ml
SLE
Healthy

Incubation of PBMCs with TLR9 agonist results in lower levels of TNF-α, IL-6, MIP-1α, and MIP-1β secretion in SLE patients compared with healthy controls

METHODOLOGY

The agonists used in these studies include an RNA-based TLR7 agonist (9) and a DNA-based TLR9 agonist (10). The donor population was comprised of healthy males (n=15, age: 23-60 years) and healthy females (n=15, age: 18-58 years). To further discern differences in TLR7- and TLR9-mediated immune responses, SLE patients (n=9, age: 29-78 years) on treatment with plaquenil/hydroxychloroquine were compared with healthy female subjects (n=10, age: 22-56 years). PBMCs (1×10^6) were incubated with TLR7 agonist (50 μg/ml) or TLR9 agonist (3 μg/ml), and cytokine induction was measured at 24 hr in supernatants using luminex/multiplex-based assays. Cytokine induction was also measured at 24 hr in supernatants of pDCs (2×10^5) incubated with TLR7 agonist (50 μg/ml) or TLR9 agonist (1 μg/ml). Activation marker upregulation on pDCs was assessed at 24 hr following incubation with TLR7 agonist (50 μg/ml) or TLR9 agonist (1 μg/ml) by flow cytometry using a cocktail of CD123-PE, CD80-FITC, and CD86-APC antibodies.

SUMMARY

Studies of immune response profiles in PBMCs and pDCs from healthy females and males with TLR7 agonist showed

❖ Higher levels of IFN-α and IP-10 induction in PBMCs from females than males.
❖ Higher levels of IL-12, IP-10, IFN-α, and TNF-α induction in pDCs from females than males.

Studies of immune response profiles in PBMCs and pDCs from healthy females and males with TLR9 agonist showed

❖ Similar cytokine responses, except lower IL-1Ra levels, in PBMCs from females than males.
❖ Similar cytokine levels in pDCs from both females and males.

PBMCs from female SLE patients showed

❖ Lower TLR7 agonist-induced secretion of IFN-α, TNF-α, and IP-10 compared with PBMCs from healthy females.
❖ Lower TLR9 agonist-induced secretion of TNF-α, IL-6, MIP-1α, and MIP-1β compared with PBMCs from healthy females.

Lower responses in SLE patients could be due to the fact that the subjects are on plaquenil, which inhibits endosomal TLR-mediated immune responses through neutralization of endosomal acidification.

REFERENCES

1. Iwasaki, A., and R. Medzhitov. 2004. *Nat. Immunol.* 5:987-995.
2. Akira, S., S. Uematsu, and O. Takeuchi. 2006. *Cell* 124:783-801.
3. Kadowaki, N., S. Ho, S. Antonenko, R. W. Malefyt, R. A. Kastelein, F. Bazan, and Y. J. Liu. 2001. *J. Exp. Med.* 194:863-869.
4. Jarrossay, D., G. Napolitani, M. Colonna, F. Sallusto, and A. Lanzavecchia. 2001. *Eur. J. Immunol.* 31:3388-3393.
5. Hemmi, H., O. Takeuchi, T. Kawai, T. Kaisho, S. Sato, H. Sanjo, M. Matsumoto, K. Hoshino, H. Wagner, K. Takeda, and S. Akira. 2000. *Nature* 408:740-745.
6. Diebold, S. S. Kaisho, T.; Hemmi, H. Akira, S. Reis e Sousa, C. 2004. *Science* 303:1529-1531.
7. Leadbetter, E. A., I. R. Rifkin, A. M. Hohlbaum, B. C. Beaudette, M. J. Shlomchik, and A. Marshak-Rothstein. 2002. *Nature* 416:603-607.
8. Savarese, E., O. W. Chae, S. Trowitzsch, G. Weber, B. Kastner, S. Akira, H. Wagner, R. M. Schmid, S. Bauer, and A. Krug. 2006. *Blood* 107:3229-3234.
9. Lan, T., M. Dai, D. Wang, F. G. Zhu, E. R. Kandimalla, and S. Agrawal. 2009. *J. Med. Chem.* 52:6871-6879.
10. Yu, D., M. R. Putta, L. Bhagat, Y. Li, F. G. Zhu, D. Wang, J. X. Tang, E. R. Kandimalla, and S. Agrawal. 2007. *J. Med. Chem.* 50:6411-6418.

www.iderapharma.com