A Novel Toll-like Receptor Antagonist for Autoimmune and Inflammatory Diseases: Safety and Pharmacodynamics in a Multiple-Dose Phase 1 Clinical Trial

Poster # 333

T. Sullivan, G. Atiee, L. Bhagat, W. Jiang, J. Murphy, N. Ostavnenko, M. Precopio, M. Putta, E. Kandimalla, R. Arbeit

Idera Pharmaceuticals, Inc., 167 Sidney Street, Cambridge, MA and Icon Development Solutions, San Antonio, TX

INTRODUCTION

IMO-3100 is a modified oligonucleotide designed as an antagonist of both TLR7 and TLR9. Immune complexes containing host-derived nucleic acids have been shown to induce inflammatory responses mediated through TLR7 and TLR9, and manifest many autoimmune diseases (1). Blocking TLR7- and TLR9-mediated immune responses through use of an antagonist represents a novel approach for the treatment of selected autoimmune diseases. IMO-3100 has been effective in reducing disease progression in preclinical models of lupus, rheumatoid arthritis, psoriasis, and hyperlipidemia (2-7). In non-human primates, IMO-3100 has shown potent inhibition of immune responses mediated through TLR7 and TLR9, but not TLR4 (8, 9). A rising single-dose clinical trial showed IMO-3100 was well tolerated and inhibited immune responses mediated through TLR7 and TLR9 (10).

The present multiple-dose Phase 1 clinical trial randomized 24 healthy subjects to receive saline, IMO-3100 at 0.32 mg/kg twice/week, or IMO-3100 at 0.64 mg/kg/week, for four weeks. The objective of trial was to study safety and antagonism of TLR7- and TLR9-mediated activity in isolated PBMCs.

Rationale of Use of Antagonist in Autoimmune Diseases

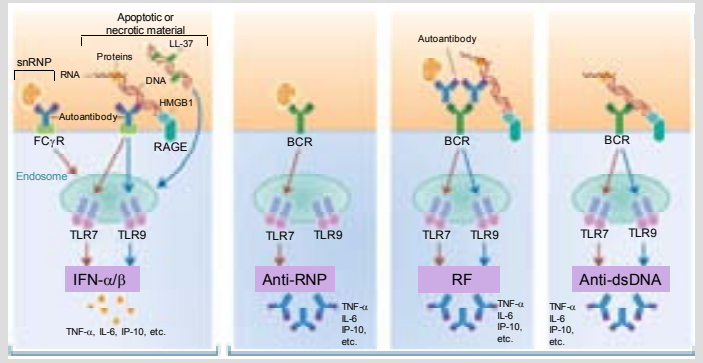

Apoptotic or necrotic material
LL-37
snRNP
Proteins
RNA
DNA
HMGB1
Autoantibody
FCγR
RAGE
Endosome
BCR
TLR7
TLR9
IFN-α/β
Anti-RNP
RF
Anti-dsDNA
TNF-α, IL-6, IP-10, etc.

pDC B cell

Design of TLR Antagonist Oligonucleotides

Synthetic oligodeoxynucleotides containing unmethylated CpG or synthetic dinucleotide motifs, referred to as immune stimulatory motifs, act as agonists of TLR9. Binding of immune stimulatory oligonucleotides to the ectodomain of TLR9 causes conformational changes in the endodomain of the receptor leading to recruitment of adaptor molecules and initiation of subsequent immune signaling pathways (11). Incorporation of 2'-O-substituted modifications adjacent to immune stimulatory motifs on the 5'-side, referred to as immune regulatory motifs, leads to antagonism of TLR9 activity through competitive binding. As a result of cross-talk between TLR9 and TLR7, we have observed that TLR9 antagonists act as TLR7 antagonists as well (12, 13).

Through structure activity relationship studies, we have created a novel class of oligonucleotide-based compounds that act as dual TLR7/TLR9 antagonists. IMO-3100 is our lead TLR7/TLR9 antagonist drug candidate.

A typical TLR7/TLR9 antagonist structure is shown below.

Immune Regulatory Motif → N; Immune Stimulatory Motif → CG*

5'-NNN**N** **CG***NNNN-3'

N = 2'-O-methylribonucleotide; **G*** = 7-deaza-dG

IMO-3100: Efficacy in Preclinical Disease Models

- Psoriasis: IL-23 and LL37 models in C57BL/6 mice
 - -Prevention and treatment models
- Rheumatoid Arthritis:
 - Bovine collagen-induced model in DBA/1 mice
 - Enhancement of etanercept activity
 - Collagen-antibody-induced model in BALB/c mice
- Lupus: MRL-lpr and NZBW/F1 mice
- Multiple Sclerosis: EAE model in SJL/j mice
- Colitis: TNBS-induced model in SJL/j mice
- Pulmonary Inflammation: OVA- and silica-induced models in BALB/c mice
- Hyperlipidemia: high-fat diet models in C57BL/6 and ApoE$^{-/-}$ mice
 - Characteristics of a chronic inflammation model

STUDY DESIGN

IMO-3100: Phase 1 Multiple Dose Clinical Trial

- Four weeks of s.c. administration in healthy subjects
 - Objectives:
 - Safety and tolerability
 - Demonstrate target engagement via ex vivo pharmacodynamic study
 - Pharmacokinetics
- Treatments (n=8 per group, randomized block design)
 - IMO-3100 0.64 mg/kg/wk
 - IMO-3100 0.32 mg/kg 2x/wk
 - Placebo (saline)
- Study schedule
 - Screening within 3 weeks prior to study drug administration
 - Admission 24 hours prior to first dose of study drug
 - Twice-weekly outpatient visits days 4-29
 - Follow-up days 36 and approx 30 days after last study drug

Schedule for injections

Day 1 4 8 11 15 18 22 25 29 36

Schedule for blood collection for ex vivo study

Injection Site Reactions

	IMO-3100 mg/kg		
Observation	**0.32**	**0.64**	**Placebo**
	n=8	n=8	n=8
Any injection site reaction	**8**	**8**	**0**
Injection site erythema	8	8	0
Injection site induration	8	8	0
Injection site pain	1	4	0
Injection site pruritus	5	2	0

Adverse Events

	IMO-3100 mg/kg		
Analyte	**0.32**	**0.64**	**Placebo**
	n=8	n=8	n=8
Subjects with any AE	**2**	**3**	**1**
Total number of AE events	**4**	**10**	**1**
Increased CK*	1	1	
Increased ALT/AST*	1	1	
Upper respiratory infection		2	
Pruritus	1	1	
Rash		1	
Earache		1	
Rhinorrhea		1	
Gastroenteritis†		1	
Urinary tract infection†		1	
Lymphadenopathy			1

* All CK and ALT/AST elevations were post-exercise and were moderate or severe

† All other AE were mild except UTI and gastroenteritis were moderate

Serum Biochemical Parameters*

		IMO-3100 mg/kg			
Analyte		**0.32**	**0.64**	**All**	**Placebo**
		n=8	n=8	n=16	n=8
Subjects with any abnormal		**8**	**7**	**15 (94%)**	**8**
CK	↑	1	1	**2 (13%)**	
ALT	↑	1	1	**2 (13%)**	
AST	↑	1	1	**2 (13%)**	
LDH	↑	1	1	**2 (13%)**	
Bicarbonate	↑	1	2	**3 (19%)**	
Calcium	↓	2		**2 (13%)**	2
Glucose	↑	2	1	**3 (19%)**	2
Leucopenia		1	1	**2 (13%)**	1
Anemia			1	**1 (6%)**	1
HCT	↓		1	**1 (6%)**	1
RBC	↓	1		**1 (6%)**	1
Chloride	↑		1	**1 (6%)**	2

* Showing all analytes outside the range

Bioanalysis of IMO-3100 in Plasma

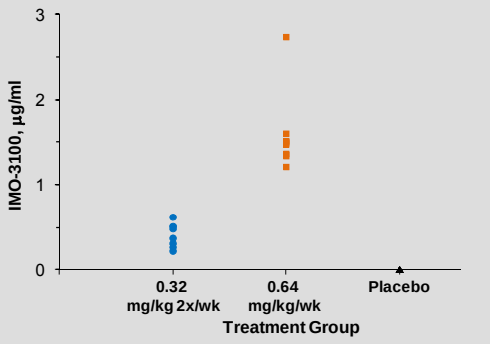

IMO-3100, μg/ml
0
1
2
3
0.32 mg/kg 2x/wk
0.64 mg/kg/wk
Placebo
Treatment Group

Plasma levels of IMO-3100 4 hours post-dose on day 1

Phase 1 trial site San Antonio, TX → Collect blood & ship overnight for each individual sample → SeraCare Gaithersburg, MD

Isolate PBMCs → Culture with TLR7 agonist / Culture with TLR9 agonist / Culture with medium alone → Measure cytokines in supernatants by multiplex

GM-CSF, IFN-α, IFN-γ, IL-1Ra, IL-1β, IL-2, IL-2R, IL-4, IL-5, IL-6, IL-7, IL-8, IL-10, IL-12, IL-13, IL-15, IP-10, MCP-1, MIG, MIP-1α, MIP-1β, RANTES, TNF-α

→ Compare TLR7- and TLR9- mediated immune responses

- IMO-3100 versus placebo*

*Fold induction of each cytokine at each time point was calculated as follows:

Fold induction = pg/ml (TLR agonist-stimulated well)/pg/ml (medium control well)

% change was calculated as follows:

% change = Fold induction (day X)/Fold induction (day 1)

Exclusions: Values for a given cytokine were excluded when Fold induction (day 1) ≤ 1. Also, outliers, defined as ≥300 % change at individual time points, were excluded in the analysis as indicated in individual cytokine plots.

RESULTS

IMO-3100: Inhibition of TLR7-Mediated Immune Responses

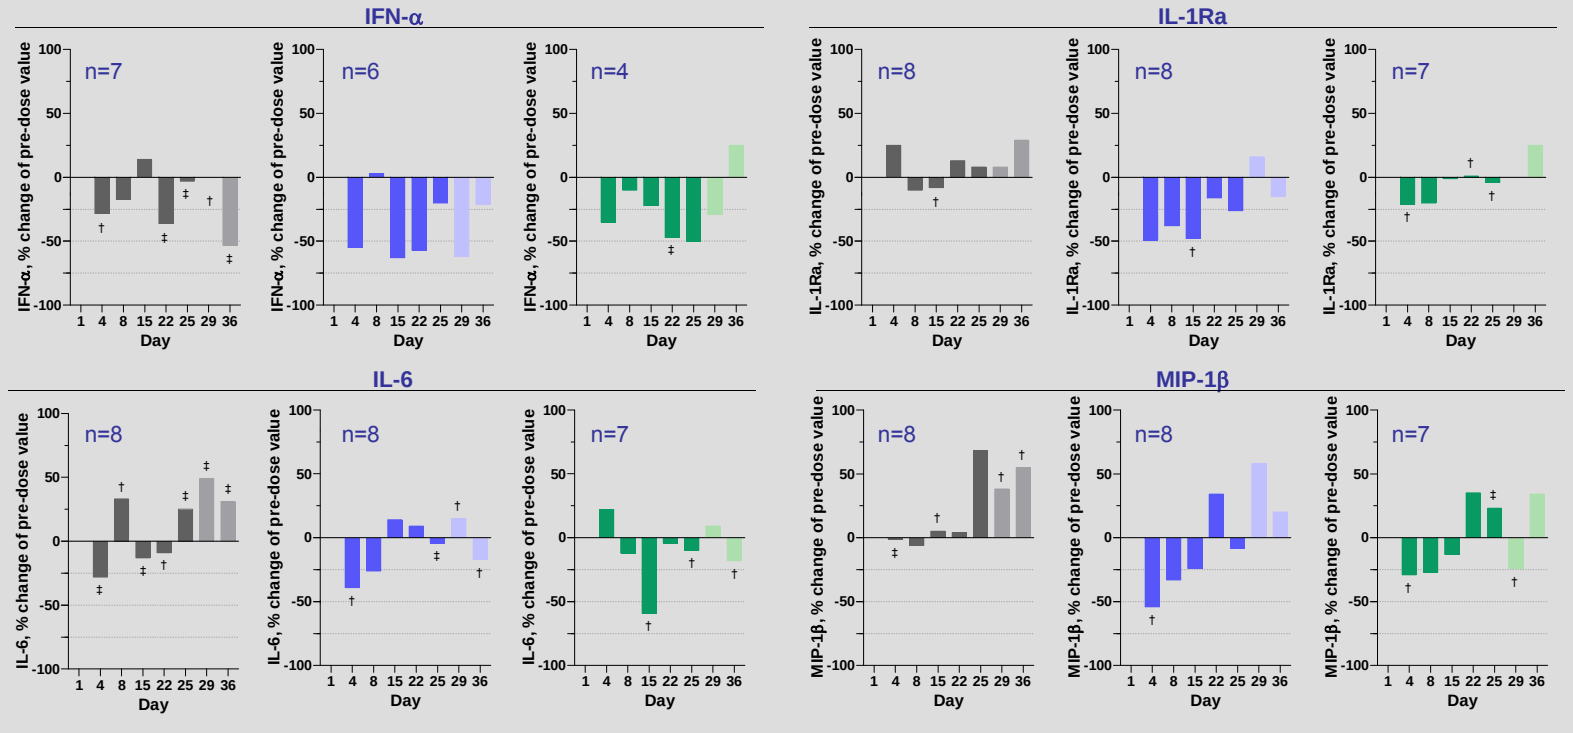

IFN-α
IL-1Ra
IL-6
MIP-1β
n=7
n=6
n=4
n=8
n=8
n=7
n=8
n=8
n=7
n=8
n=8
n=7
IFN-α, % change of pre-dose value
IL-1Ra, % change of pre-dose value
IL-6, % change of pre-dose value
MIP-1β, % change of pre-dose value
1 4 8 15 22 25 29 36
Day

IMO-3100: Inhibition of TLR9-Mediated Immune Responses

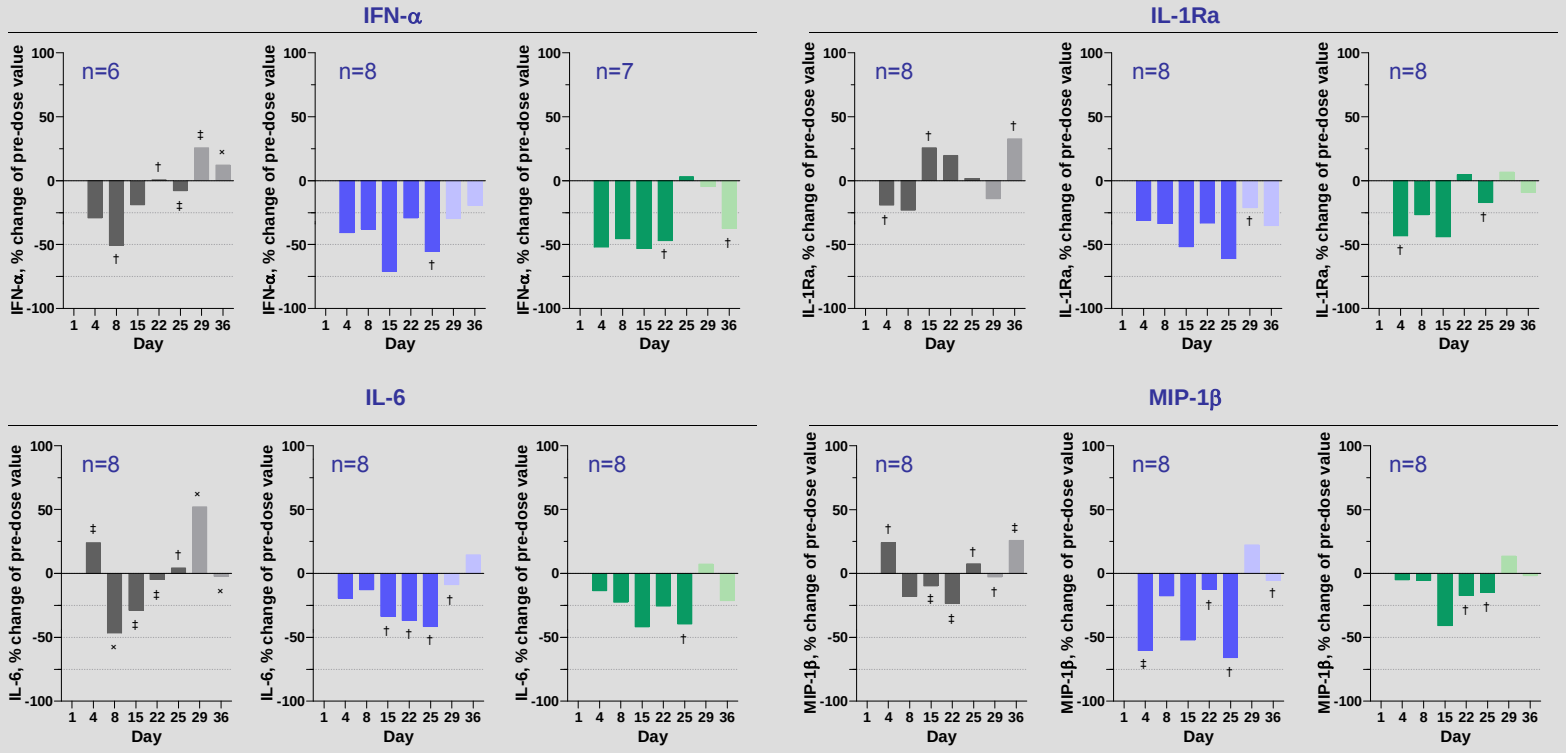

IFN-α
IL-1Ra
IL-6
MIP-1β
n=6
n=8
n=7
n=8
n=8
n=8
n=8
n=8
n=8
n=8
n=8
n=8
IFN-α, % change of pre-dose value
IL-1Ra, % change of pre-dose value
IL-6, % change of pre-dose value
MIP-1β, % change of pre-dose value
1 4 8 15 22 25 29 36
Day

Lighter shaded bars indicate timepoints following the last dose. ■ Placebo ■ 0.32 mg/kg/twice wk ■ 0.64mg/kg/wk

† One, ‡ two, or × three outliers excluded at given time point

SUMMARY

- Safety Evaluation
 - No serious adverse events (SAEs)
 - No drug-related discontinuations
 - Most common drug-related AEs were mild-moderate injection site reactions
 - No episodes of fever, chills, flu-like symptoms
- Pharmacokinetic Evaluation
 - Dose-proportional
 - No evidence of accumulation (data from day 22 not shown)
- Proof of Target Engagement
 - Sustained inhibition of TLR7- and TLR9-mediated cytokine induction in PBMCs isolated after IMO-3100 treatment
 - Kinetics of inhibition vary between different cytokines
 - PBMCs from placebo-treated subjects showed no consistent differences in post-dose cytokine responses
 - Model successfully demonstrates pharmacodynamic effect

REFERENCES

1. Rifkin, I.R., et al. (2005) Immunol. Rev. **204**, 27-42.
2. Zhu, F.G., et al. (2007) Keystone Symposium on Biology of B Cells in Health and Disease. Banff, Alberta, Canada, February 6-12.
3. Zhu, F.G., et al. (2008) FOCIS Annual Meeting, Boston, MA, June 5-9.
4. Zhu, F.G., et al. (2008) 4th Annual Meeting of the Oligonucleotide Therapeutics Society, Boston, MA, October 15-18.
5. Bhagat, L., et al. (2008) 4th Annual Meeting of the Oligonucleotide Therapeutic Society, Boston, MA, October 15-18.
6. Jiang, W., et al. (2009) 2009 Annual Meeting of The American Association of Immunologists, Seattle, WA, May 8-12.
7. Zhu, F.G., et al. (2009). Annual Meeting of American College of Rheumatologists (ACR) 2009, Philadelphia, PA, October 17-21.
8. Bhagat, L., et al. (2010) Keystone Symposium on Tolerance and Autoimmunity, Taos, NM, February 21-26.
9. Bhagat, L., et al. (2010) ACR/ARHP Annual Meeting 2010, Atlanta, GA; November 06-11.
10. Sullivan, T., et al. (2010) 6th Annual Meeting of the Oligonucleotide Therapeutic Society, Dana Point, CA, October 20-23.
11. Latz , E., et al. (2007) Nat. Immunol. **8**, 772-779.
12. Wang, D., et al. (2009) J. Med. Chem. **52**, 551-558.
13. Yu, D., et al. (2009) J. Med. Chem. **52**, 5108-5114.

www.iderapharma.com