Poster #473

Novel class of DNA-based compounds act as antagonists for TLR7 and 9: In vitro and in vivo studies in MRL-lpr and NZBW/F1 mouse models

Fu-Gang Zhu, Ekambar R. Kandimalla, Dong Yu, Daqing Wang, and Sudhir Agrawal

Idera Pharmaceuticals, Inc, 345 Vassar Street, Cambridge, MA 02139, USA.

Introduction

- Toll-like receptors (TLR) recognize pathogen-associated molecular patterns (PAMP) and induce pathogen specific innate and adaptive immune responses. Of the eleven TLRs (TLR1-11) known in mammals, TLR3 (double-stranded viral and synthetic RNA), TLR7/8 (single-stranded viral and synthetic RNA, and ribonucleosides), TLR9 (unmethylated CpG motif containing bacterial and synthetic DNA) recognize nucleic acid molecular patterns.
- Recent studies suggest involvement of TLRs in autoimmune diseases in animals and human subjects with susceptible background.
- Our structure-activity relationship studies of DNA have led us to identify a novel class of DNA-based compounds, which act as antagonists of TLR9 and 7[1-3].
- Our earlier studies showed that antagonist candidates inhibit activity of TLR9 and 7 agonists in vitro and in vivo in mice and human cell-based assays.
- In the present study we have evaluated the effects of novel antagonist candidates in lupus prone MRL-lpr and NZBW/F1 mouse models.

1. Bhagat L et al. presented at 93rd American Association of Immunologist (AAI) annual meeting, Boston, MA, May 12-16, 2006.
2. Wang D et al. presented at 2nd Annual Meeting of the Oligonucleotide Therapeutics Society, New York, NY, October 19 – 21, 2006.
3. Wang D et al. presented at the American College of Rheumatology (ACR) annual meeting, Washington, DC, November 10-15, 2006.

In vivo immune stimulation of an antagonist and an agonist in mice

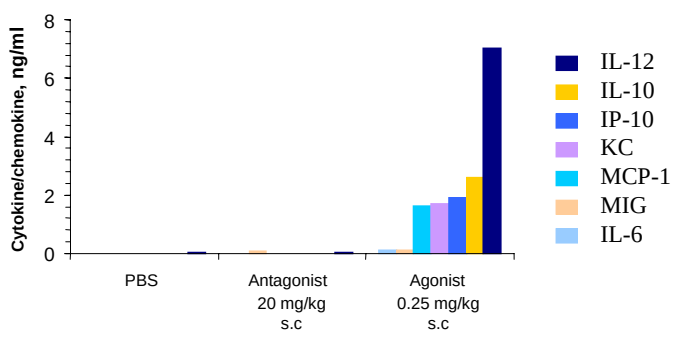

Cytokine/chemokine, ng/ml
0
2
4
6
8
PBS
Antagonist 20 mg/kg s.c
Agonist 0.25 mg/kg s.c
IL-12
IL-10
IP-10
KC
MCP-1
MIG
IL-6

TLR antagonist: Sustained inhibitory effect on agonist of TLR9 induced IL-12 *in vivo*

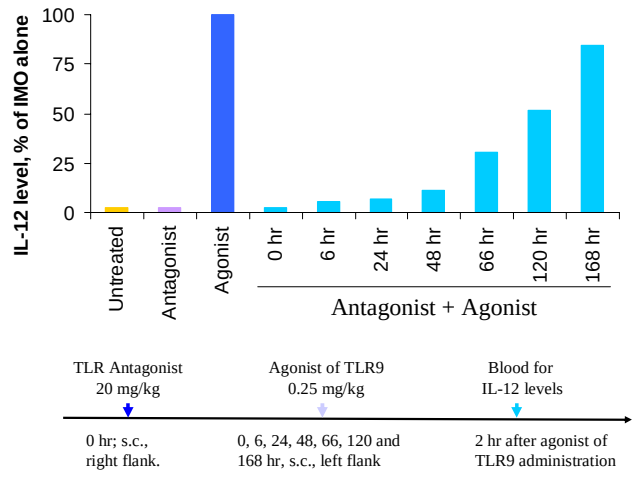

IL-12 level, % of IMO alone
0
25
50
75
100
Untreated
Antagonist
Agonist
0 hr
6 hr
24 hr
48 hr
66 hr
120 hr
168 hr
Antagonist + Agonist
TLR Antagonist 20 mg/kg
Agonist of TLR9 0.25 mg/kg
Blood for IL-12 levels
0 hr; s.c., right flank.
0, 6, 24, 48, 66, 120 and 168 hr, s.c., left flank
2 hr after agonist of TLR9 administration

TLR antagonist: Specificity of inhibitory activity

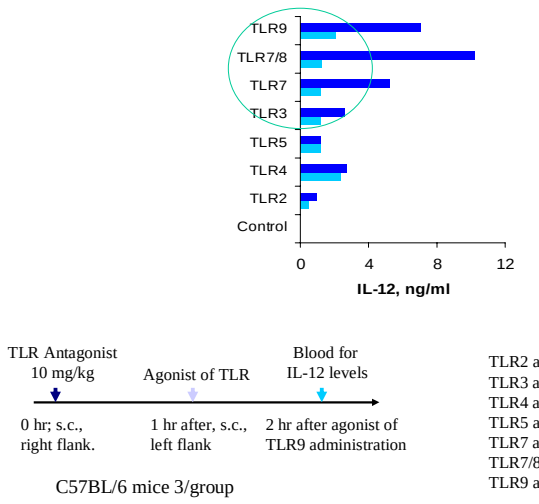

TLR9
TLR7/8
TLR7
TLR3
TLR5
TLR4
TLR2
Control
0
4
8
12
IL-12, ng/ml
TLR Antagonist 10 mg/kg
Agonist of TLR
Blood for IL-12 levels
0 hr; s.c., right flank.
1 hr after, s.c., left flank
2 hr after agonist of TLR9 administration

C57BL/6 mice 3/group

TLR2 agonist – MALP2 (0.5 mg/kg)
TLR3 agonist – polyI.polyC (20 mg/kg)
TLR4 agonist – LPS (0.25 mg/kg)
TLR5 agonist – Flagellin (0.25 mg/kg)
TLR7 agonist – Loxoribine (100 mg/kg)
TLR7/8 agonist – R848 (0.1 mg/kg)
TLR9 agonist – IMO (0.25 mg/kg)

Effect of an antagonist on NZBW/F1 mouse spleen B cell cytokine secretion

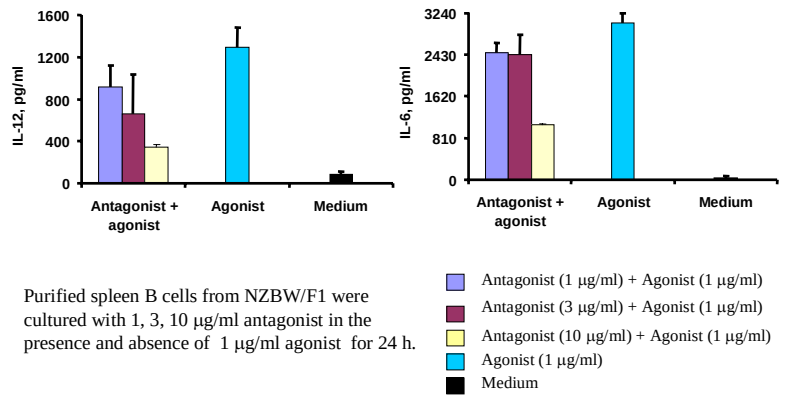

IL-12, pg/ml
0
400
800
1200
1600
IL-6, pg/ml
0
810
1620
2430
3240
Antagonist + agonist
Agonist
Medium
Antagonist (1 μg/ml) + Agonist (1 μg/ml)
Antagonist (3 μg/ml) + Agonist (1 μg/ml)
Antagonist (10 μg/ml) + Agonist (1 μg/ml)
Agonist (1 μg/ml)
Medium

Purified spleen B cells from NZBW/F1 were cultured with 1, 3, 10 μg/ml antagonist in the presence and absence of 1 μg/ml agonist for 24 h.

Effect of an antagonist on mouse B lymphocyte proliferation

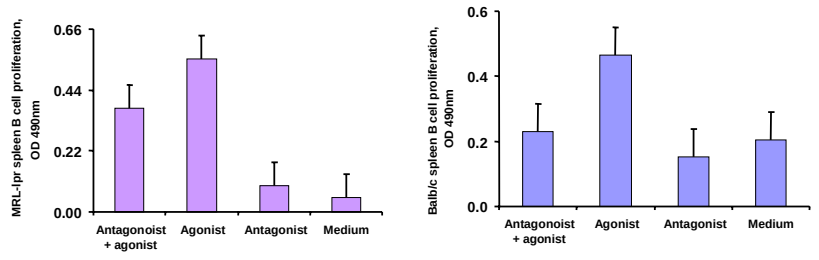

MRL-lpr spleen B cell proliferation, OD 490nm
0.00
0.22
0.44
0.66
Balb/c spleen B cell proliferation, OD 490nm
0.0
0.2
0.4
0.6
Antagonoist + agonist
Agonist
Antagonist
Medium

Purified spleen B cells from MRL-lpr and Balb/c mice were cultured with 1 μg/ml antagonist in the presence and absence of 0.3 μg/ml agonist for 72 h.

NZBW/F1 mouse studies - Experimental Protocol

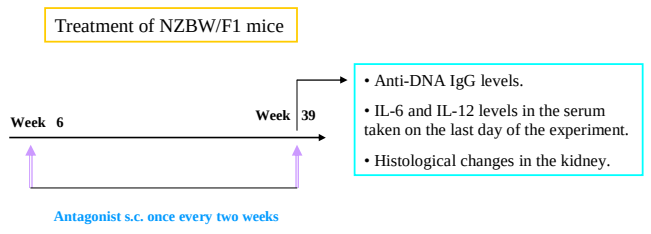

Treatment of NZBW/F1 mice
Week 6
Week 39
• Anti-DNA IgG levels.
• IL-6 and IL-12 levels in the serum taken on the last day of the experiment.
• Histological changes in the kidney.
Antagonist s.c. once every two weeks

N=5

Groups:

1. **PBS**
2. **Antagonist candidate 1: 300 μg (10 mg/kg)**
3. **Antagonist candidate 2: 300 μg (10 mg/kg)**

The long term effect of antagonist candidates on serum cytokine levels in NZBW/F1 mice

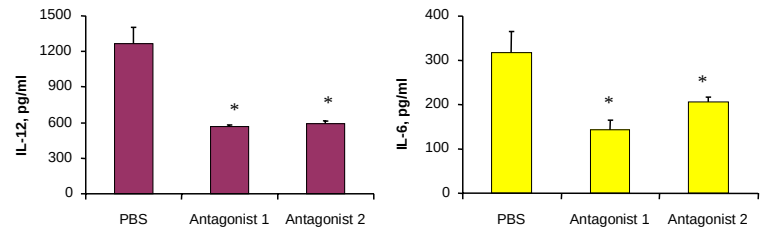

IL-12, pg/ml
0
300
600
900
1200
1500
IL-6, pg/ml
0
100
200
300
400
PBS
Antagonist 1
Antagonist 2

* = P < 0.05

Serum IL-6 and IL-12 levels on the last day of the experiment (week 39).

Serum anti-DNA IgG levels in NZBW/F1 mice

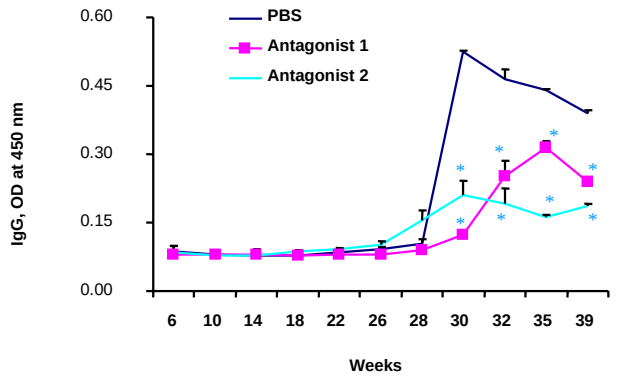

PBS
Antagonist 1
Antagonist 2
IgG, OD at 450 nm
0.00
0.15
0.30
0.45
0.60
6
10
14
18
22
26
28
30
32
35
39
Weeks

Serum anti-DNA total IgG levels were significantly lower in mice treated with antagonist 1 or 2 than in PBS treated mice (* = p values < 0.05).

Effect of antagonist candidates on renal disease development in NZBW/F1 mice

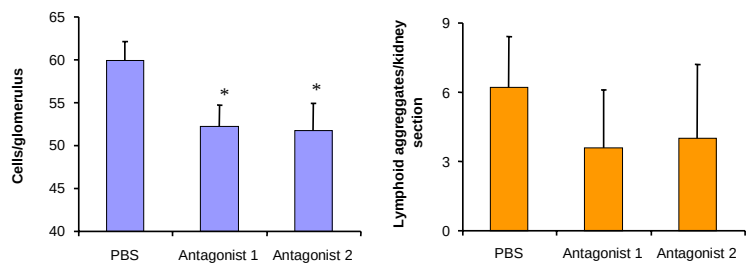

Cells/glomerulus
40
45
50
55
60
65
Lymphoid aggregates/kidney section
0
3
6
9
PBS
Antagonist 1
Antagonist 2

* = P < 0.05

Kidneys were examined for histological changes at the end of experiment (week 39).

MRL-lpr mouse studies - Experimental Protocol

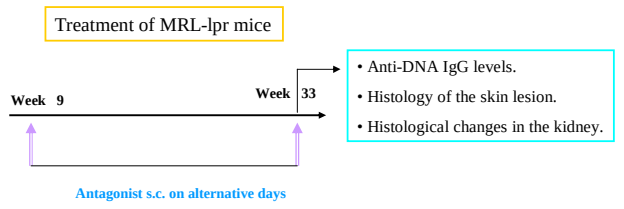

Treatment of MRL-lpr mice
Week 9
Week 33
• Anti-DNA IgG levels.
• Histology of the skin lesion.
• Histological changes in the kidney.
Antagonist s.c. on alternative days

N=5

Groups:

1. **PBS**
2. **Antagonist candidate 1: 40 μg (1.3 mg/kg)**
3. **Antagonist candidate 1: 100 μg (3.3 mg/kg)**

Serum anti-DNA IgG levels in MRL-lpr mice

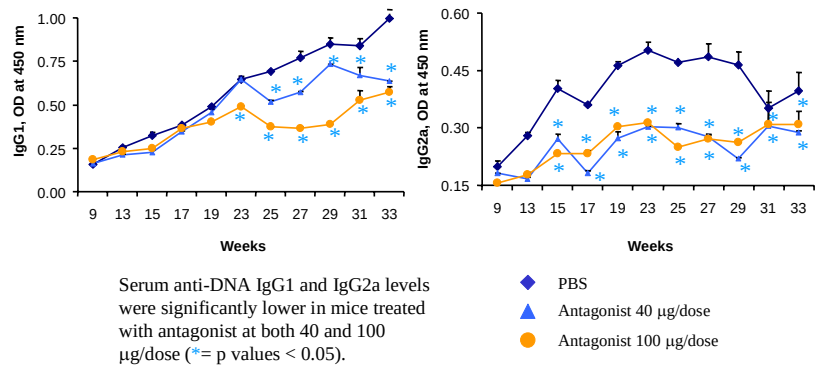

IgG1, OD at 450 nm
0.00
0.25
0.50
0.75
1.00
IgG2a, OD at 450 nm
0.15
0.30
0.45
0.60
9
13
15
17
19
23
25
27
29
31
33
Weeks
PBS
Antagonist 40 μg/dose
Antagonist 100 μg/dose

Serum anti-DNA IgG1 and IgG2a levels were significantly lower in mice treated with antagonist at both 40 and 100 μg/dose (*= p values < 0.05).

Effect of antagonist treatment on the development of skin lesions in MRL-lpr mice

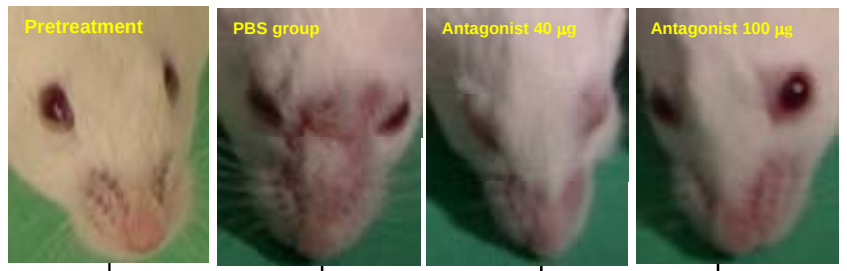

Pretreatment
PBS group
Antagonist 40 μg
Antagonist 100 μg

Pretreatment at week 8

4 out of 5 mice developed skin lesions at week 27

None of the mice developed skin lesions treated groups at week 27

Effect of an antagonist candidate on the development of skin lesions in MRL-lpr mice (histological changes)

Epidermal hyperplasia in the facial skin at the site of lesion

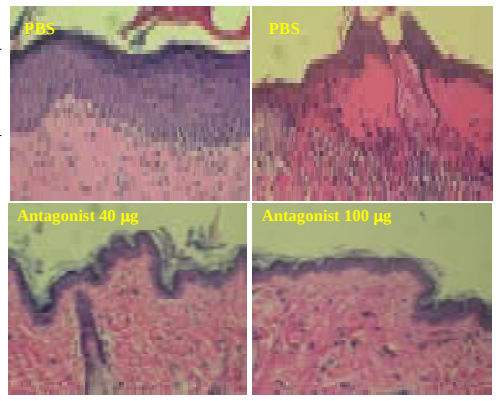

PBS
PBS
Antagonist 40 μg
Antagonist 100 μg

Ulceration and inflammation in the dorsum skin at the site of lesion

Extensive epidermal hyperplasia, ulceration and inflammation were observed in the skin sections of all five mice in PBS treated group. None of the skin sections of the mice treated with antagonist at 40 or 100 μg dose showed pathological changes observed in untreated mice.

Effect of an antagonist candidate on renal disease development in MRL-lpr mice

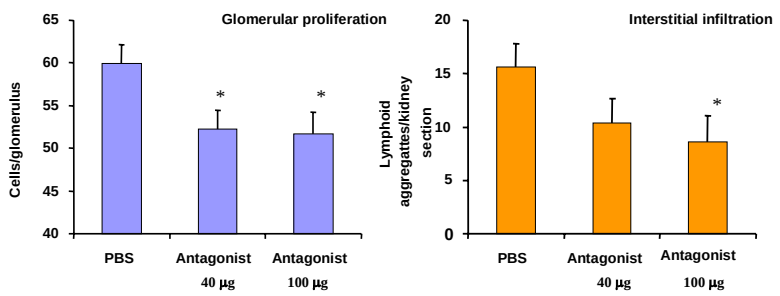

Glomerular proliferation
Cells/glomerulus
40
45
50
55
60
65
Interstitial infiltration
Lymphoid aggregates/kidney section
0
5
10
15
20
PBS
Antagonist 40 μg
Antagonist 100 μg

* = P < 0.05

Kidneys were examined for histological changes at the end of experiment (week 33).

Effect of an antagonist candidate on inflammation and glomerular proliferation in the kidney of MRL-lpr mice

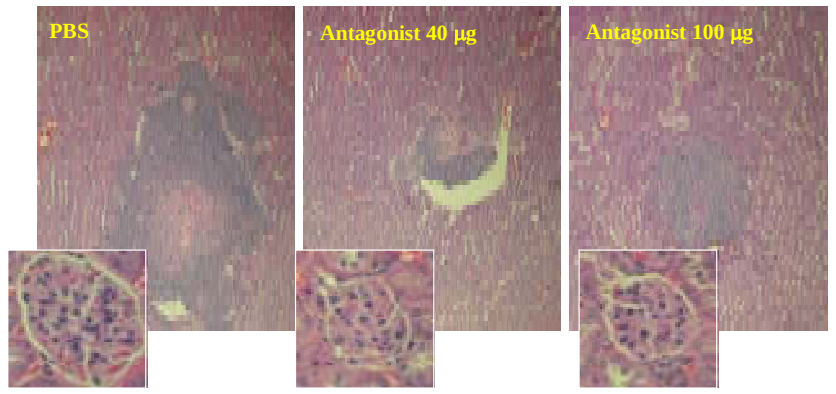

PBS
Antagonist 40 μg
Antagonist 100 μg

- Large lymphoid aggregates are present in the kidney tissues of PBS treated mice compared with antagonist treated mice at either dose (magnification= 100).
- Increased cell numbers observed in a representative glomerulus of the kidneys of PBS treated mice compared with antagonist treated mice (see inserts, magnification= 1000).

Conclusions

- A novel class of synthetic DNA-based antagonist candidates has been identified.
- Antagonist candidates have shown to block immune stimulation by agonists of TLR9 and 7 in preclinical studies.
- Evaluation of antagonist candidates in MRL-lpr and NZBW/F1 mouse models shows that
 - Antagonist treatment is effective in preventing development of butterfly skin rash in MRL-lpr mice.
 - Antagonists are effective in reducing serum anti-DNA antibody levels in both MRL-lpr and NZBW/F1 mice.
 - MRL-lpr and NZBW/F1mice treated with antagonists showed improved renal histopathology.
- TLR antagonists show potential as novel candidates for the treatment of lupus and other autoimmune diseases.