IMO-8400, an Antagonist of Toll-like Receptors 7, 8, and 9, in Development for Genetically Defined B-cell Lymphomas: Safety and Tolerability in Phase 1 and Phase 2 Clinical Trials

Robert D. Arbeit, Tim Sullivan, and Lou Brenner | Idera Pharmaceuticals, Inc., 167 Sidney Street, Cambridge, MA 02139

BACKGROUND

Toll-like Receptors Have Central Role in the Innate Immune System

- Toll-like receptors (TLRs) are pathogen-associated molecular pattern recognition receptors of the innate immune system
- TLRs 7 and 9, which are expressed in human B-cells, respond to DNA- and RNA-based ligands by initiating a signaling cascade mediated through NF-κB, IRAK1/4, BTK, and JAK/STAT
- DNA- and RNA-based ligands for TLRs 7 and 9 also are generated as damage-associated molecular patterns in certain autoimmune diseases and malignancies

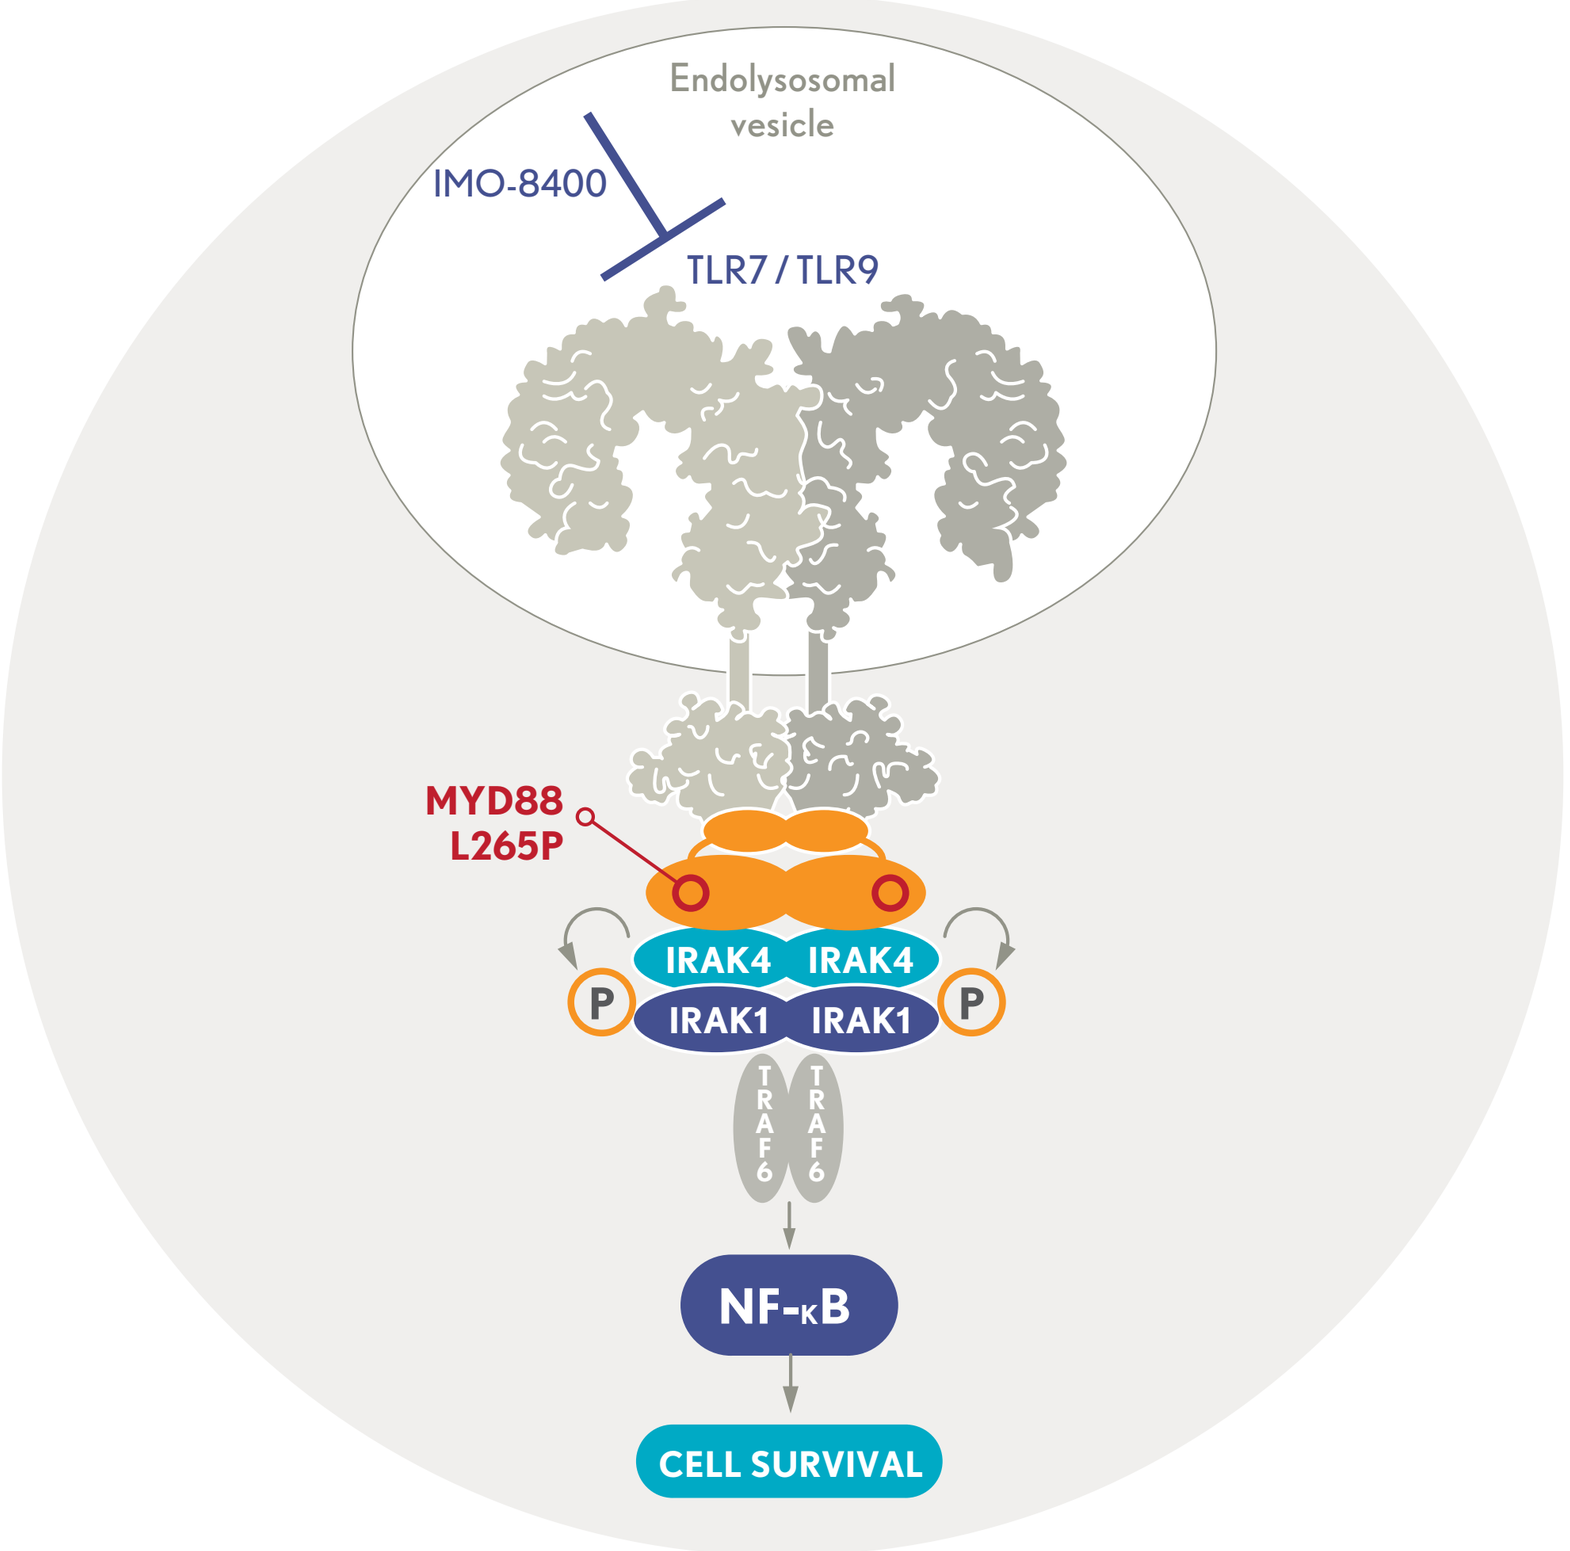
Endolysosomal vesicle
IMO-8400
TLR7 / TLR9
MYD88 L265P
IRAK4 IRAK4
IRAK1 IRAK1
P
P
TRAF6
TRAF6
NF-κB
CELL SURVIVAL

MYD88 L265P Oncogenic Mutation Over-activates TLR Signaling

- MYD88 is a key adaptor molecule in TLR signaling
- Recently, the oncogenic mutation MYD88 L265P has been described and reported to occur in patients with activated B-cell–like diffuse large B-cell lymphoma (ABC-DLBCL), Waldenström's macroglobulinemia (WM), and other B-cell malignancies[1]
- MYD88 L265P drives over-expression of TLR 7 and 9 signaling, creating an anti-apoptotic microenvironment that promotes cancer cell survival and proliferation
- Recent epidemiological data confirm a strong association between the presence of MYD88 L265P and poor outcomes in patients with DLBCL[2]
- Specific targeting of signaling initiated by TLRs 7 and 9 is therefore a novel approach to the treatment of B-cell lymphomas harboring the MYD88 L265P mutation

Ongoing Clinical Development of IMO-8400 for Genetically Defined Forms of B-cell Lymphoma

- Preclinical data presented at AACR 2014, including from xenograft tumor studies, showed that IMO-8400 inhibited cell growth and signaling and reduced tumor growth in WM and DLBCL models harboring the MYD88 L265P mutation[3]
- Based on the accumulated preclinical data, clinical safety, and demonstrated mechanism of action of IMO-8400, Phase 1/2 clinical trials of IMO-8400 are being conducted in both relapsed/refractory patients with WM and in patients with DLBCL and the MYD88 L265P mutation

REFERENCES:
1. Lim et al. AACR 2013 Abstract #2332.
2. Fernandez-Rodriguez et al. *Leukemia*. 2014, 28:2104-6.
3. Bhagat et al. AACR 2014 Abstract #2570.

CLINICAL TRIAL RESULTS

Phase 1 Clinical Trial in Healthy Volunteers (Study 8400-001)

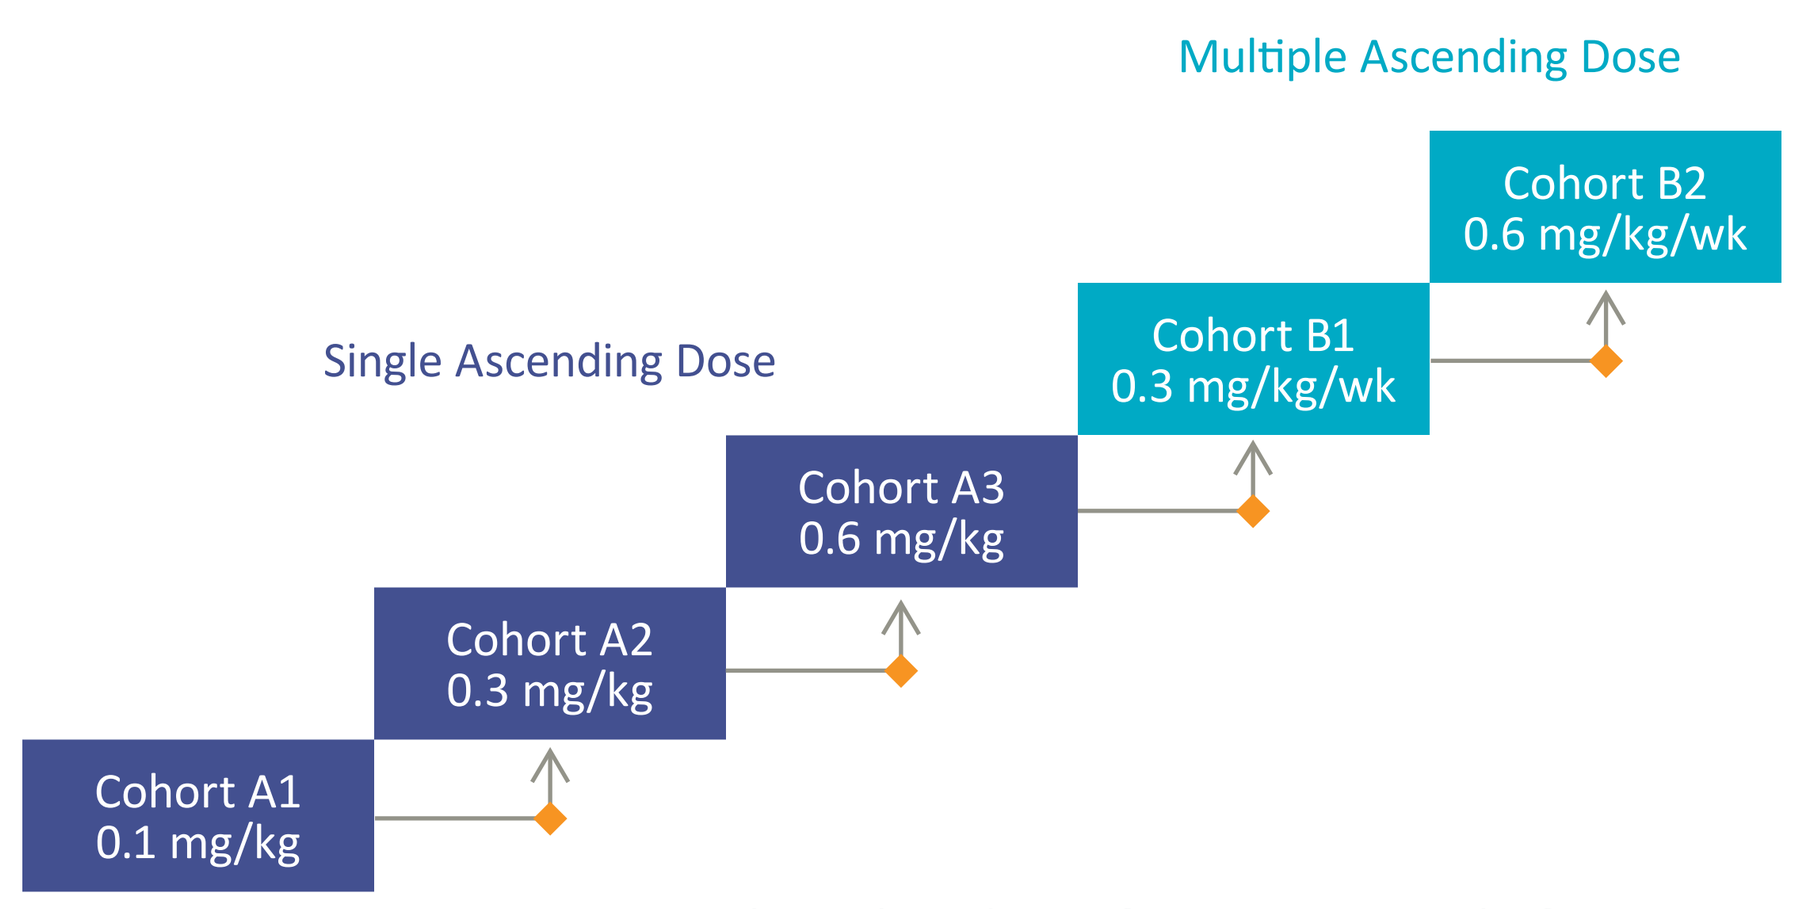
Multiple Ascending Dose
Single Ascending Dose
Cohort B2 0.6 mg/kg/wk
Cohort B1 0.3 mg/kg/wk
Cohort A3 0.6 mg/kg
Cohort A2 0.3 mg/kg
Cohort A1 0.1 mg/kg

IMO-8400 (n)	Placebo (n)
6	3
6	3
6	3
6	3
6	–

◆ Dose escalation based on safety review 1 week after treatment.

Phase 1 Safety Results Excluding Injection-Site Reactions

Event	Single Dose (mg/kg)				Multiple Dose (mg/kg/wk)		
	0.1 (n=6)	0.3 (n=6)	0.6 (n=6)	PLA (n=6)	0.3 (n=6)	0.6 (n=6)	PLA (n=6)
Any adverse event	1 (16%)	2 (33%)	0	1 (16%)	2 (33%)	1 (16%)	2 (33%)
Respiratory tract infection	1	1	–	1	1	–	1
Dizziness	–	1	–	–	2	1	2
Itching	–	–	–	–	1	–	–
Toothache	–	–	–	–	–	1	–
Headache	–	–	–	–	–	–	1
Neck muscle pain	–	–	–	–	–	–	1

No patterns of treatment-related changes in safety laboratory tests.

Summary of Injection-site Reactions in Phase 1 Trial

Injection-Site Reaction	Single Dose (mg/kg)			Multiple Dose (mg/kg/wk)	
	0.1 (n=6)	0.3 (n=6)	0.6 (n=6)	0.3 (n=6)	0.6 (n=6)
Erythema	2 (33%)	3 (50%)	2 (33%)	1 (16%)	5 (84%)
Induration	–	–	–	1	1
Tenderness	–	–	1	1	–
Pruritus	–	–	–	–	2

At all ISR assessments, no blistering, ulceration, or necrosis was observed.

Phase 2 Clinical Trial in Patients with Psoriasis (Study 8400-201)

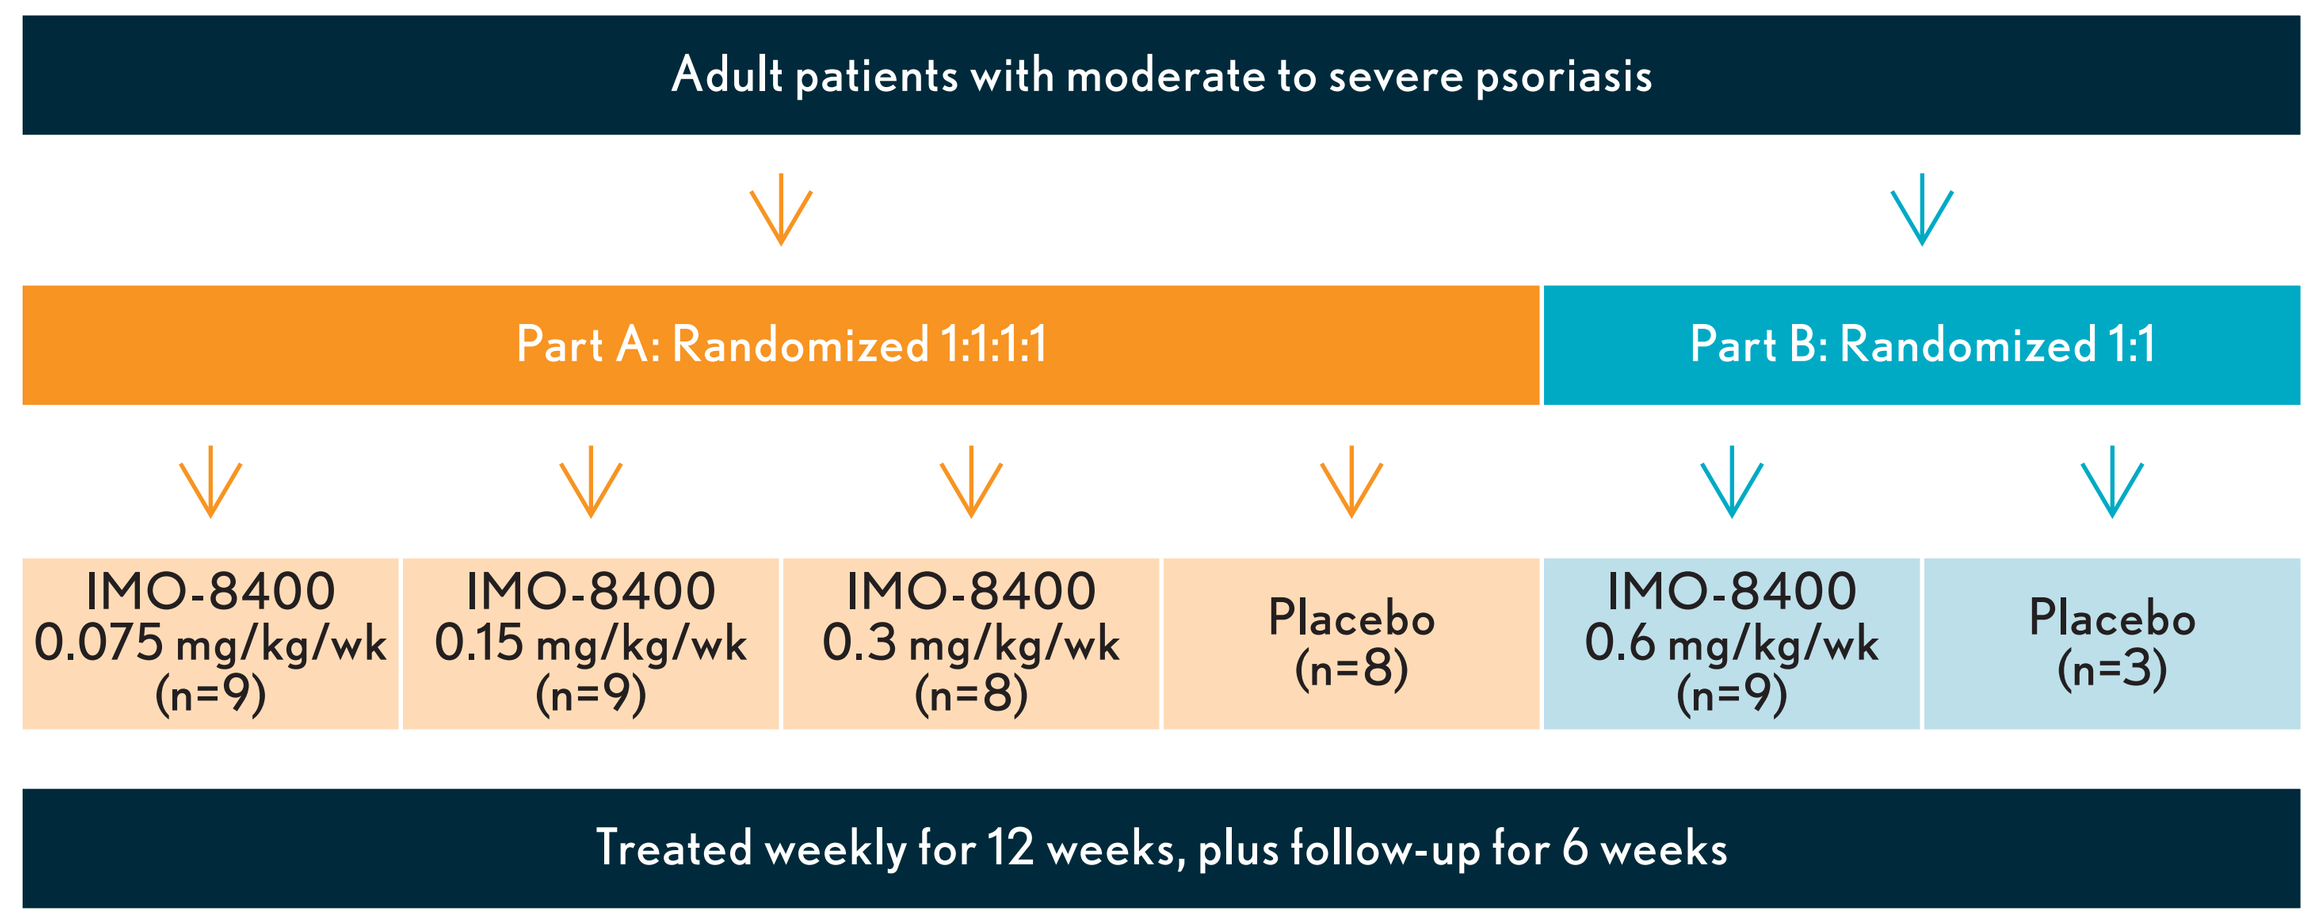
Adult patients with moderate to severe psoriasis
Part A: Randomized 1:1:1:1
Part B: Randomized 1:1
IMO-8400 0.075 mg/kg/wk (n=9)
IMO-8400 0.15 mg/kg/wk (n=9)
IMO-8400 0.3 mg/kg/wk (n=8)
Placebo (n=8)
IMO-8400 0.6 mg/kg/wk (n=9)
Placebo (n=3)
Treated weekly for 12 weeks, plus follow-up for 6 weeks

Phase 2 Safety Population and Exposure

Patient Disposition	IMO-8400 mg/kg/week					Placebo
	0.075	0.15	0.30	0.60	All	
Safety Population	9	9	8	9	35	11
Exposure <6 weeks	1	2	–	–	3	–
Exposure 7-11 weeks	1	–	2	–	3	2
Exposure 12 weeks	7	7	6	9	29	9

Phase 2 Adverse Events by System Organ Class and Relatedness

System Organ Class (≥2 events)	All IMO-8400	All IMO-8400–Related	All Placebo	All Placebo-Related
	(n=35)		(n=11)	
Any Event	30 (86)	23 (66)	9 (82)	6 (54)
Gastrointestinal Disorders	10 (29)	9 (26)	1 (9)	–
General Disorders (excl. ISR)	14 (40)	17 (49)	2 (18)	1 (9)
Infections And Infestations	6 (17)	0	1 (9)	–
Musculoskeletal Disorders	7 (20)	1 (3)	5 (46)	2 (18)
Nervous System Disorders	15 (43)	5 (14)	5 (46)	–
Psychiatric Disorders	4 (11)	0	1 (9)	–
Renal and Urinary Disorders	2 (6)	2 (6)	1 (9)	–
Respiratory Disorders	4 (11)	0	2 (18)	–
Skin Disorders	3 (9)	1 (3)	3 (27)	2 (18)

Phase 2 Adverse Events by Preferred Term and Relatedness

Preferred Term (≥1 events)	All IMO-8400	All IMO-8400–Related	All Placebo	All Placebo-Related
	(n=35)		(n=11)	
Any Event	30 (86)	23 (66)	9 (82)	6 (54)
Abdominal Discomfort	2 (6)	1 (3)	1 (9)	–
Diarrhea	7 (20)	6 (17)	–	–
Nausea	3 (9)	3 (9)	–	–
Vomiting	2 (6)	2 (6)	–	–
Fatigue	6 (17)	6 (17)	1 (9)	–
Influenza-like Illness	7 (20)	6 (17)	1 (9)	1 (9)
Muscle Spasms	–	–	2 (18)	2 (18)
Somnolence	3 (9)	3 (9)	–	–
Insomnia	3 (9)	–	1 (9)	–
Polyuria	2 (6)	2 (6)	–	–

Phase 2 Selected Laboratory Parameters

- AST – Treatment-emergent elevations
 - 3/11 (27%) Placebo
 - 6/35 (17%) IMO-8400 (similar at all dose levels)
 - All of the elevations were Grade 1
 - Most were <1.5x ULN and transient; the maximum was 2.2x ULN
- Hemoglobin – Treatment-emergent decreases
 - 1/11 (9%) Placebo
 - 4/35 (11%) IMO-8400 (one at each dose level)
 - All of the decreases were Grade 1; the minimum was 11.6 g/dL
- Neutrophils – Treatment-emergent decreases
 - No treatment-emergent neutropenia was seen in any patient
- Platelets – Treatment-emergent decreases
 - 1/11 (9%) Placebo – 101,000 at baseline, nadir 87,000
 - 5/35 (14%) IMO-8400
 - All Grade 1; the minimum was 137,000

Summary of Phase 2 Safety Results

- There were no treatment-related SAEs, no treatment-related severe AEs, and no discontinuations due to treatment-related AEs
- There were 3 events reported as severe (Grade 3), none of which were assessed as treatment-related. All three subjects discontinued study treatment after the event
- All other events were assessed as mild or moderate in severity
- Allowing for the limited number of placebo subjects, the overall frequency and distribution of events by treatment group was similar
- The frequency of gastrointestinal events was numerically higher among IMO-8400-treated patients; the frequencies of musculoskeletal and skin events were higher among placebo patients
- Events assessed as possibly or probably related to study drug showed a similar distribution
- No abnormalities of laboratory results were attributed to study drug. Analysis of the primary data by numerical values and by CTC grading showed no differences between the patients treated with IMO-8400 or placebo

Summary of Injection-site Reactions in Phase 2 Trial

- Each prior injection site was formally evaluated for reactions at every visit, representing 114 scheduled site-evaluations per patient, or >1000 per treatment group of 9 patients
- One placebo subject had a single reaction of mild erythema and induration, which was presumably trauma-related
- Among the IMO-8400 subjects there was a dose-related increase in the frequency of ISRs, being uncommon and transient at dose levels of ≤0.15 mg/kg, and increasingly prevalent at the higher dose levels
- The most common ISR was erythema
 - Erythema was seen at least transiently with most of the injections administered in the 0.6 mg/kg dose group
- Induration, pruritus, and tenderness were rare at the lower dose levels, and experienced at least once by about half the patients in higher dose groups, typically for brief duration
 - Pain was reported uncommonly at any dose level
- All but two ISRs events were assessed as mild
 - One patient at 0.3 mg/kg reported brief, moderate tenderness
 - One subject at 0.6 mg/kg reported brief, moderate pruritus
- No blistering, ulceration, or necrosis was observed in any subject

Ongoing Phase 1/2 Clinical Trial in Waldenström's Macroglobulinemia (Study 8400-401)

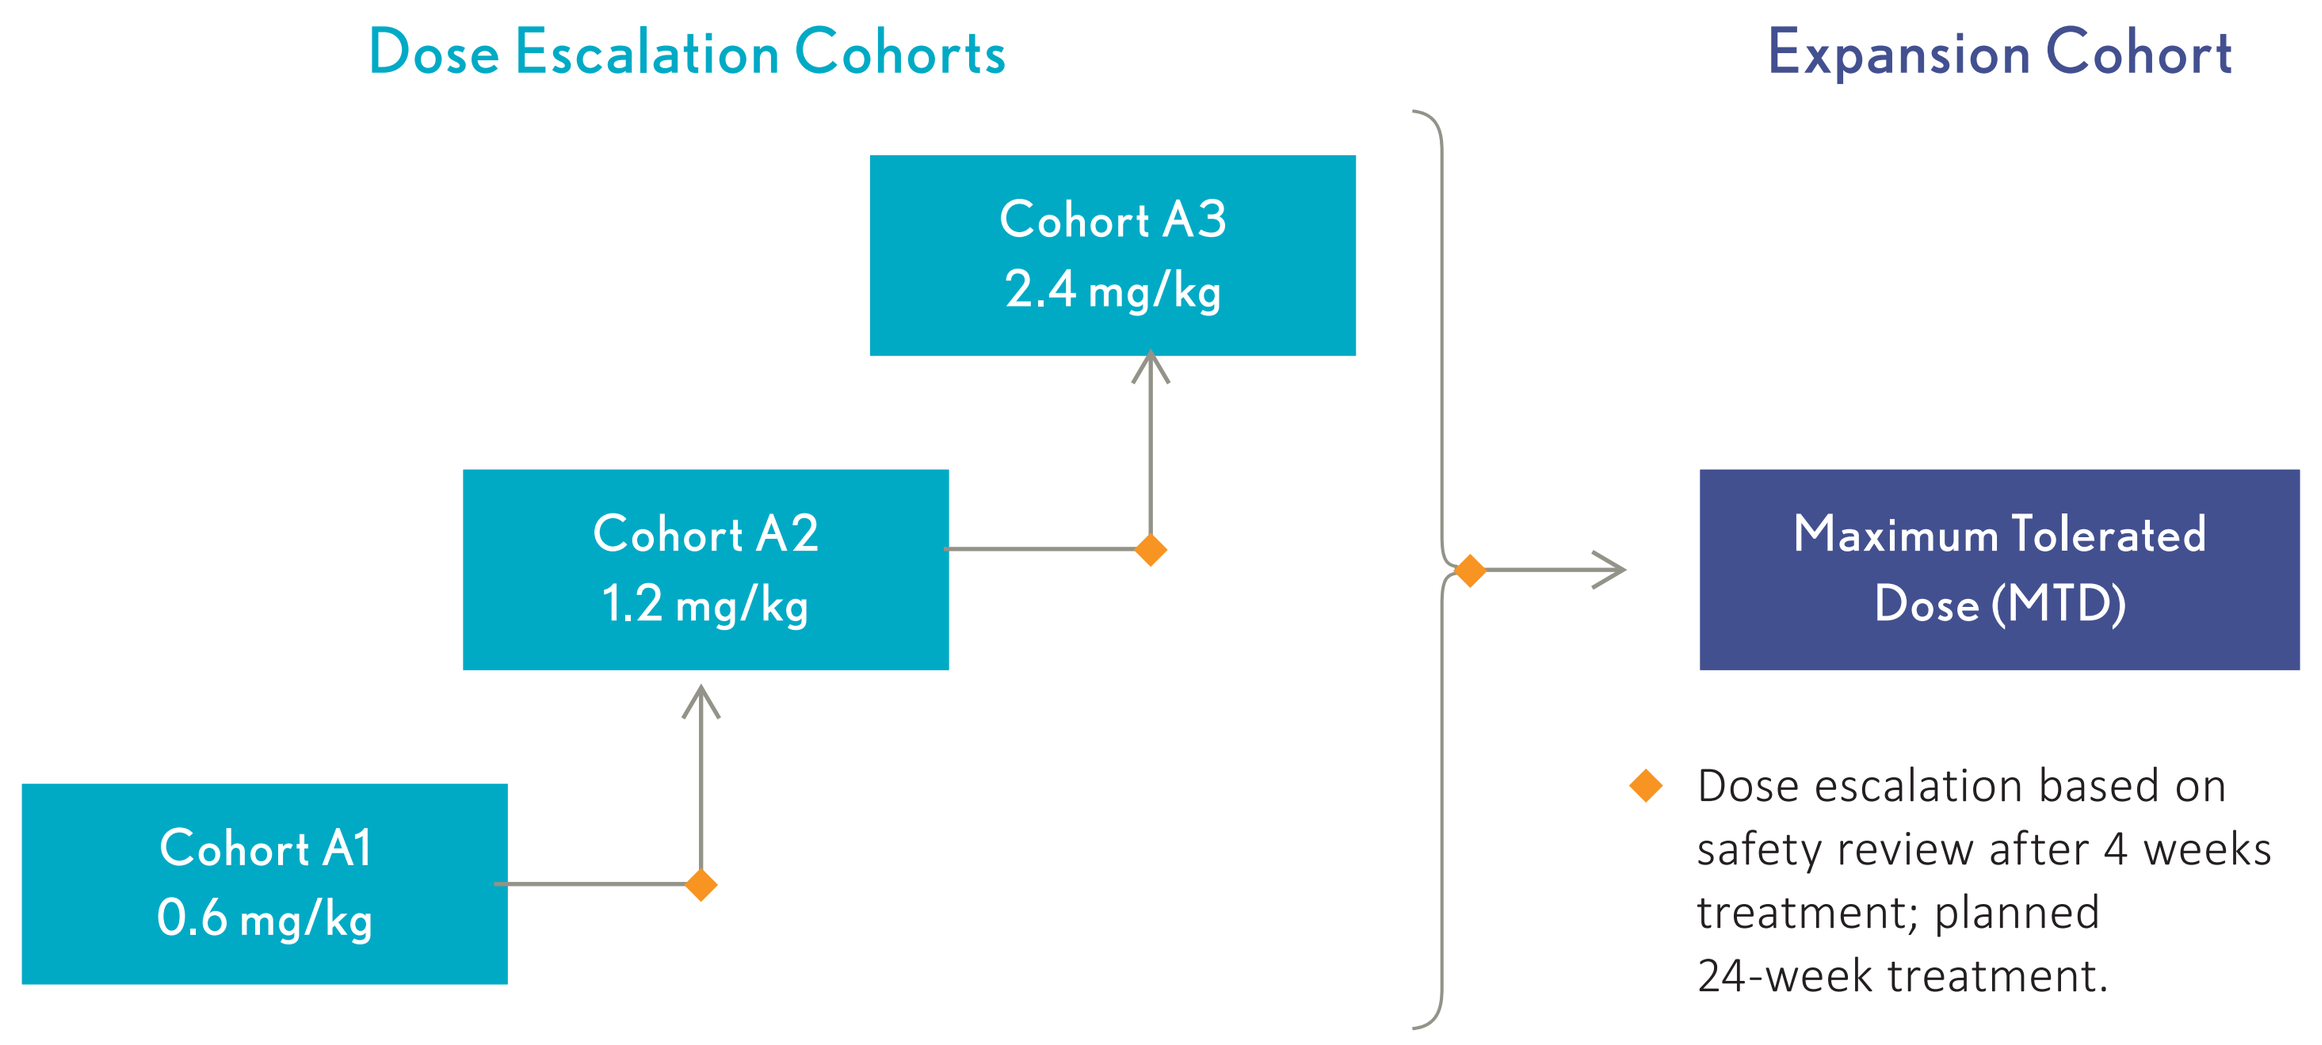
Dose Escalation Cohorts
Expansion Cohort
Cohort A3 2.4 mg/kg
Cohort A2 1.2 mg/kg
Cohort A1 0.6 mg/kg
Maximum Tolerated Dose (MTD)
Dose escalation based on safety review after 4 weeks treatment; planned 24-week treatment.

- Dose escalation status
 - Cohort 1: 0.6 mg/kg/week – dose escalation approved
 - Cohort 2: 1.2 mg/kg/week – dose escalation review planned Dec 2014
 - Cohort 3: 2.4 mg/kg/week – if approved by Data Review Committee
- Experience to date
 - No SAEs
 - No treatment-related discontinuations
- Detailed 24-week safety and clinical activity data anticipated in second half of 2015

SUMMARY

- IMO-8400 has been administered by SC injection
 - to 18 subjects as single doses of 0.1 to 0.6 mg/kg
 - to 12 subjects as 4 doses of 0.3 or 0.6 mg/kg once weekly
 - to 35 subjects as up to 12 doses of 0.075 to 0.6 mg/kg once weekly
- Across this experience, there have been
 - no treatment-related SAEs
 - no treatment-related severe AEs
 - no discontinuations due to treatment-related AEs
- The most common treatment-related AEs have been mild ISRs of erythema and induration. There have been no instances of ulceration or necrosis
- There has been no pattern of systemic AEs and no pattern of laboratory changes, including LFTs and hematology parameters
- IMO-8400 represents a novel treatment for B-cell malignancies with the MYD88 L265P mutation. It has been well-tolerated in completed clinical trials to date

Disclosure: Authors are employees of Idera Pharmaceuticals and hold stock in the company.

iderapharma.com