Phase 1b Trial of ABBV-368 + Tilsotolimod in Combination With Nab-Paclitaxel and/or Budigalimab (ABBV-181) in Patients With Recurrent/Metastatic Head and Neck Squamous Cell Carcinoma

Poster #2195

Xiuning Le,[1] Iris Gluck,[2] Corinne Maurice-Dror,[3] Aru Panwar,[4] Kathryn Gold,[5] Tong Dai,[6] Jaspreet Grewal,[7] Misako Nagasaka,[8] Ari Rosenberg,[9] Missak Haigentz, Jr,[10] Christophe Le Tourneau,[11] Irene Moreno,[12] Marc Oliva,[13] Maulik Patel,[14] Daniel Da Costa,[14] Stacie Lambert,[14] Yan Li,[14] Martha Blaney,[14] Michael McDevitt,[14] and Maura Gillison[1]

[1]Department of Thoracic Head and Neck Medical Oncology, The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Department of Radiotherapy, Sheba Medical Center, HaMerkaz, Israel; [3]Division of Oncology, Rambam Health Care Campus, Haifa, Israel; [4]Department of Head and Neck Surgical Oncology, Nebraska Methodist Hospital, Omaha, NE, USA; [5]University of California – San Diego Moores Cancer Center, La Jolla, CA, USA; [6]Department of Medical Oncology, Roswell Park Comprehensive Cancer Center, Buffalo, NY, USA; [7]Department of Medical Oncology, Norton Cancer Institute, Louisville, KY, USA; [8]Department of Oncology, Karmanos Cancer Institute, Detroit, MI, USA; [9]Department of Hematology and Oncology, University of Chicago, Chicago, IL, USA; [10]Atlantic Health System Cancer Care, Morristown, NJ, USA; [11]Department of Drug Development and Innovation (D3i), Institute Curie, Paris, France; [12]Hospital Universidad Madrid Sanchinarro, Madrid, Spain; [13]Catalan Institute of Oncology (ICO) L'Hospitalet, Barcelona, Spain; and [14]AbbVie, Inc., Redwood City, CA, USA

Poster presented at the European Society for Medical Oncology (ESMO) 2020 Congress • September 17, 2020

BACKGROUND

- Head and neck cancers account for approximately 4% of all cancers in the United States,[1] and patients with recurrent/metastatic head and neck squamous cell carcinoma (R/M HNSCC) have a poor prognosis, with a median survival in the second line of less than 7 months[2]
- Stimulation of the immune system to generate antitumor response represents a compelling therapeutic strategy, especially in patients with poor prognosis and limited treatment options
- OX40 is a type I transmembrane cell surface member of the tumor necrosis factor (TNF) receptor superfamily that plays a critical role in the enhancement of nascent immune responses and suppresses regulatory T-cell activity
- Preclinical studies have shown that TLR9 agonists can induce OX40 expression on CD4 T-cells in the tumor microenvironment; an agonistic anti-OX40 antibody can then be used to co-stimulate a tumor antigen specific T-cell immune response. This approach has led to tumor regression in mouse models[3]
- **ABBV-368** is a novel, immunoglobulin G1 anti-OX40 monoclonal antibody agonist that elicits tumor responses by stimulating effector T cells and inhibiting regulatory T cells[4]

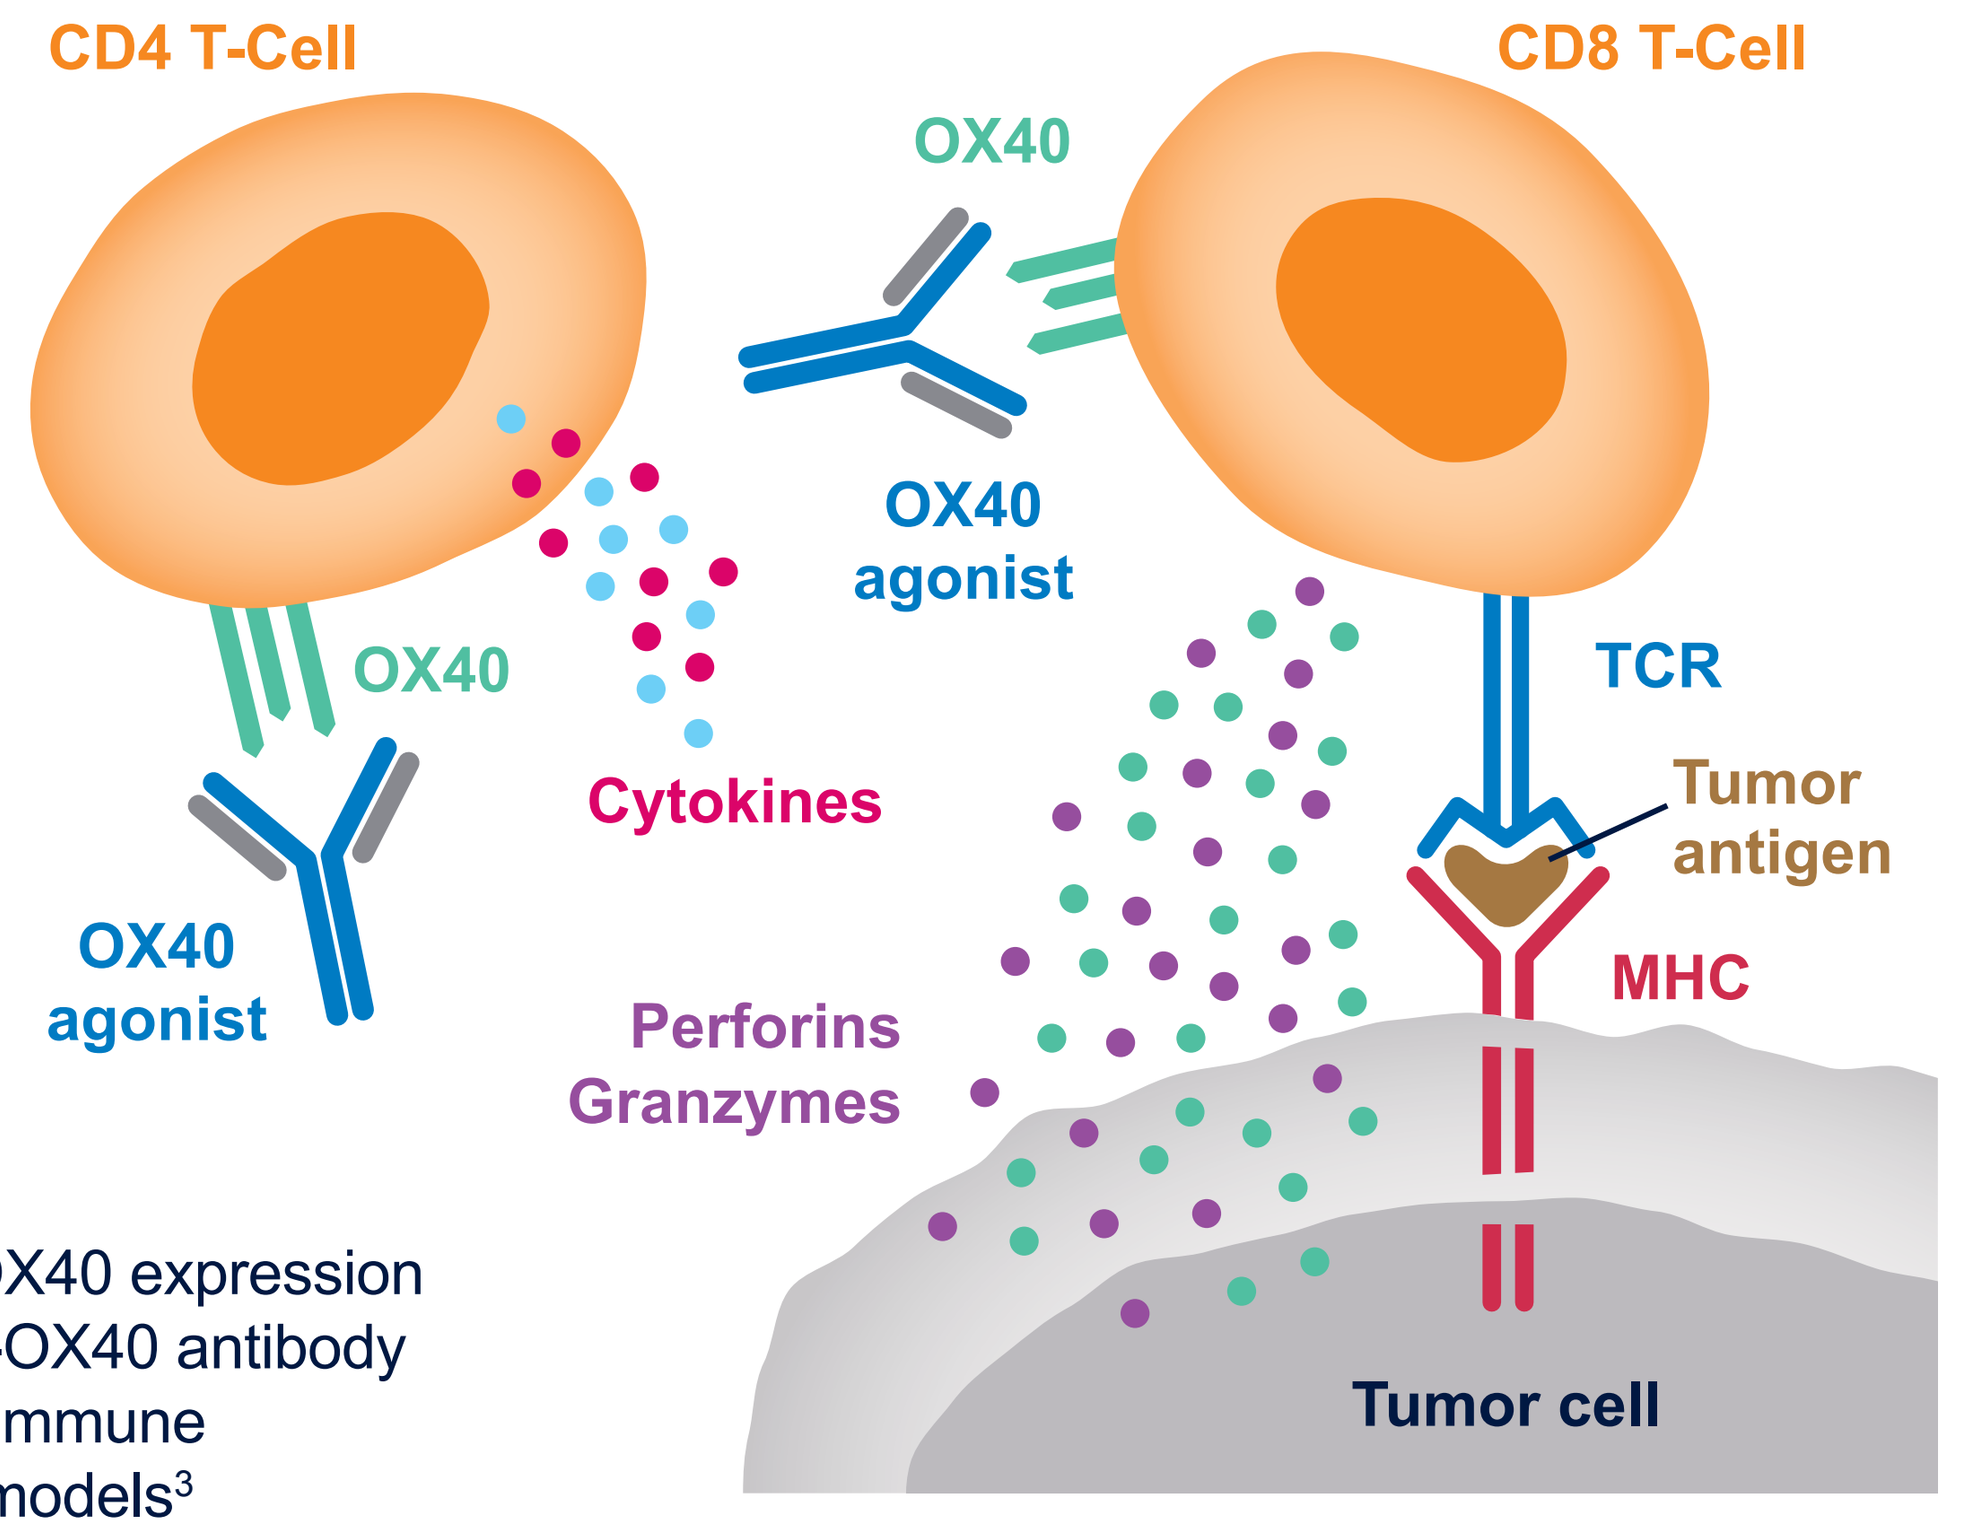
CD4 T-Cell
CD8 T-Cell
OX40
OX40 agonist
OX40
OX40 agonist
Cytokines
TCR
Tumor antigen
MHC
Perforins Granzymes
Tumor cell

MHC, major histocompatibility complex; TCR, T-cell receptor.

- **Tilsotolimod** is a synthetic toll-like receptor 9 (TLR9) agonist with immunostimulatory activity[5]
- **Budigalimab** (ABBV-181) is a humanized, recombinant immunoglobulin G1 monoclonal antibody targeting programmed cell death protein-1 (PD-1) modified to reduce Fc receptor interactions and limit effector function

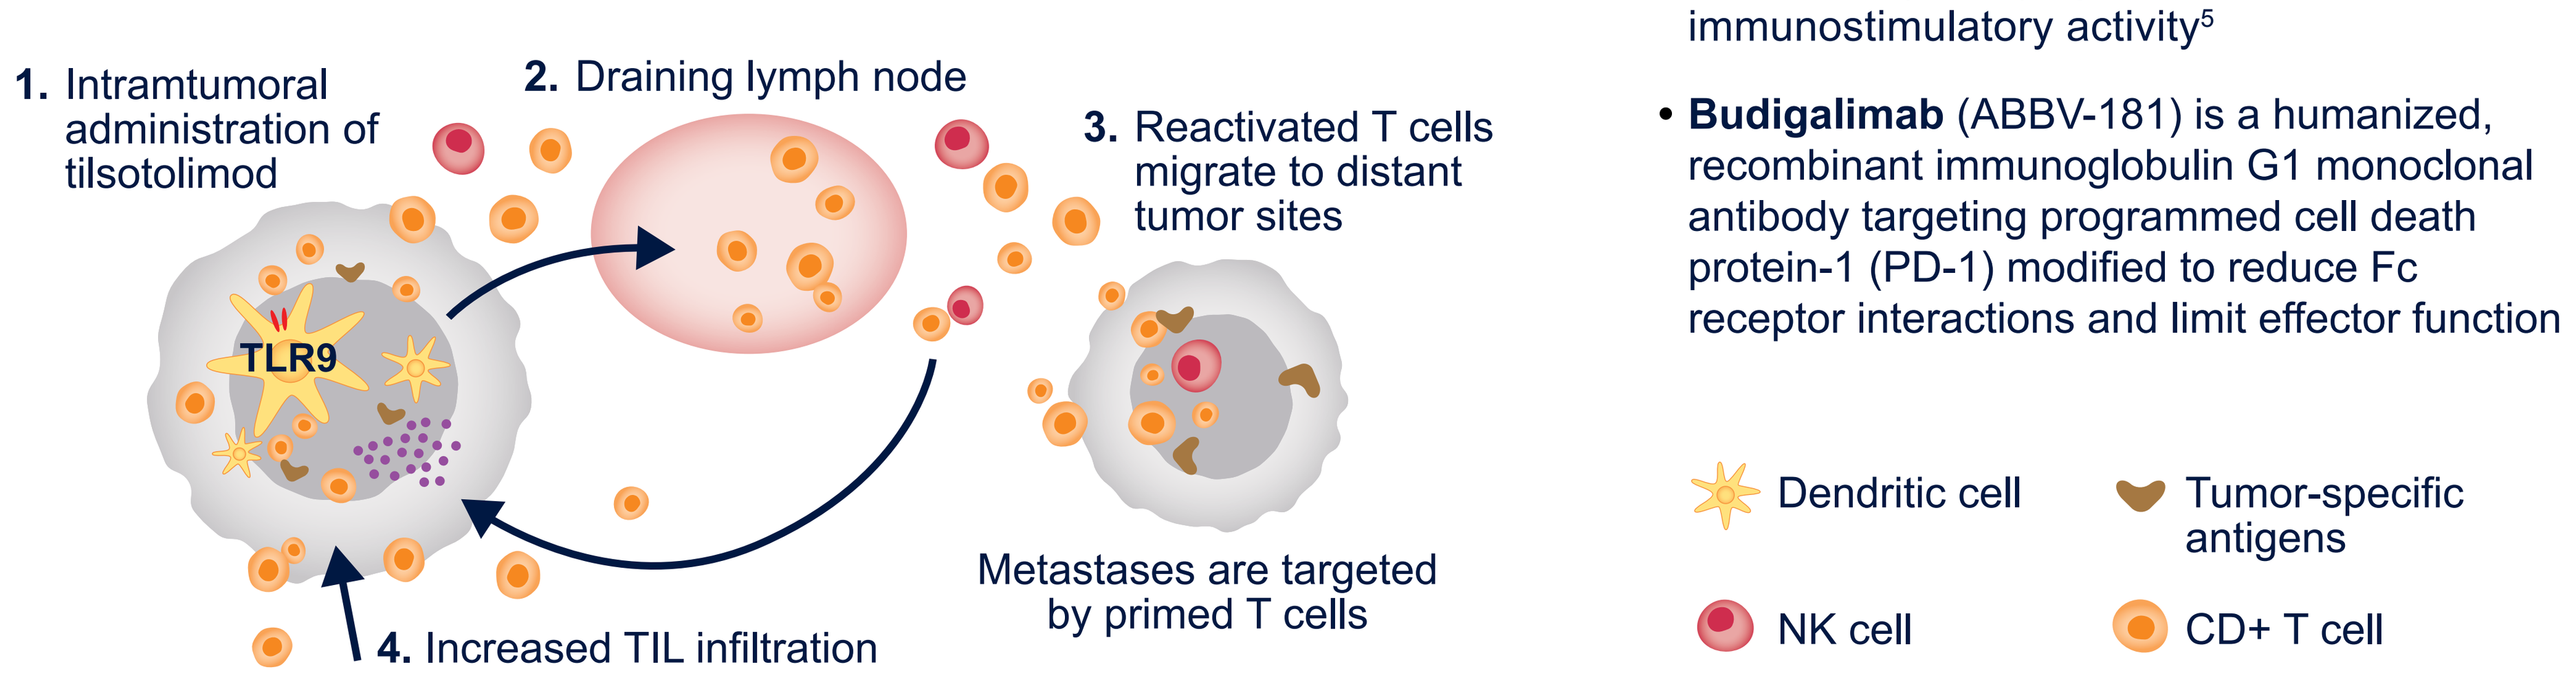
1. Intramtumoral administration of tilsotolimod
2. Draining lymph node
3. Reactivated T cells migrate to distant tumor sites
TLR9
Metastases are targeted by primed T cells
4. Increased TIL infiltration
Dendritic cell
Tumor-specific antigens
NK cell
CD+ T cell

NK, natural killer; TIL, tumor-infiltrating lymphocyte.

STUDY OBJECTIVES

1 PRIMARY

- To assess the safety, tolerability, and pharmacokinetics (PK) of ABBV-368 plus tilsotolimod; ABBV-368 plus tilsotolimod and nab-paclitaxel; and ABBV-368 plus tilsotolimod, nab-paclitaxel, and budigalimab (ABBV-181) in patients with R/M HNSCC

2 SECONDARY

- To assess the preliminary antitumor activity of ABBV-368 plus tilsotolimod; ABBV-368 plus tilsotolimod and nab-paclitaxel; and ABBV-368 plus tilsotolimod, nab-paclitaxel, and budigalimab in patients with R/M HNSCC based on Response Evaluation Criteria in Solid Tumors version 1.1 (RECIST v1.1)

EXPLORATORY

- To assess the preliminary antitumor activity of ABBV-368 plus tilsotolimod; ABBV-368 plus tilsotolimod and nab-paclitaxel; and ABBV-368 plus tilsotolimod, nab-paclitaxel, and budigalimab in patients with R/M HNSCC based on immune RECIST (iRECIST)
- To evaluate pharmacodynamic and predictive biomarkers for association with PK, safety, and efficacy measures in patients with R/M HNSCC
- To evaluate the immunogenicity of ABBV-368, tilsotolimod, and budigalimab in patients with R/M HNSCC

STUDY DESIGN

Multicenter and International

1 2 3
3 Treatment Arms

69
Estimated enrollment: **69** patients

P1b
Phase 1b

R/M HNSCC

NCT04196283
Initiated on January 22, 2020

KEY PATIENT ELIGIBILITY CRITERIA

KEY INCLUSION CRITERIA

- ✓ Age ≥18 years and weight >35 kg
- ✓ Eastern Cooperative Oncology Group performance status of 0 or 1 and life expectancy of ≥3 months
- ✓ Histologically or cytologically confirmed R/M HNSCC of the oral cavity, oropharynx, larynx, or hypopharynx that previously progressed either during or after ≤3 prior treatment regimens administered in the recurrent or metastatic setting
- ✓ Must have received 1 immunotherapy regimen that included a PD-(L)1 inhibitor
- ✓ Must have ≥1 measurable target lesion per RECIST v1.1
- ✓ Must have ≥1 lesion accessible for intratumoral (IT) injection, and lesion(s) must be ≥2 cm in longest diameter
- ✓ Adequate liver and bone marrow function

KEY EXCLUSION CRITERIA

- × Received prior treatment with OX40 or TLR agonists (excluding topical agents)
- × Uncontrolled metastases to the central nervous system
 - – Patients with brain metastases are eligible provided that there is evidence of clinically and radiographically stable disease for at least 4 weeks after therapy is administered and patients have not used prohibited levels of steroids for at least 4 weeks prior to first dose of the study drug
- × History of clinically significant uncontrolled condition(s) including but not limited to the following: uncontrolled hypertension; Grade ≥3 peripheral neuropathy; active uncontrolled infection; symptomatic congestive heart failure; unstable angina pectoris or cardiac arrhythmia; and psychiatric illness/social situation that would limit compliance with the study
- × Active autoimmune disease requiring therapy within the previous 2 years, with exceptions of vitiligo, type I diabetes mellitus, hypothyroidism, and psoriasis
- × History of inflammatory bowel disease, pneumonitis, primary immunodeficiency, bone marrow transplantation, chronic lymphocytic leukemia, solid organ transplantation, interstitial lung disease, or active or latent tuberculosis without completion of standard-of-care treatment
- × Positive test results for HIV or chronic or active hepatitis A, B, or C
 - – Patients who have a history of hepatitis B or C and undetectable levels of hepatitis B viral DNA or hepatitis C viral RNA after antiviral therapy may be enrolled
- × Received a live attenuated vaccine within 4 weeks of the first study dose

STUDY TREATMENT

THIS STUDY CONSISTS OF 3 TREATMENT ARMS:

1. Patients in **Arm 1** will receive intravenous (IV) ABBV-368 + tilsotolimod IT
2. Patients in **Arm 2** will receive IV ABBV-368 + tilsotolimod IT + IV nab-paclitaxel
3. Patients in **Arm 3** will receive IV ABBV-368 + tilsotolimod IT + IV nab-paclitaxel + IV budigalimab

Figure 1 – Study Design

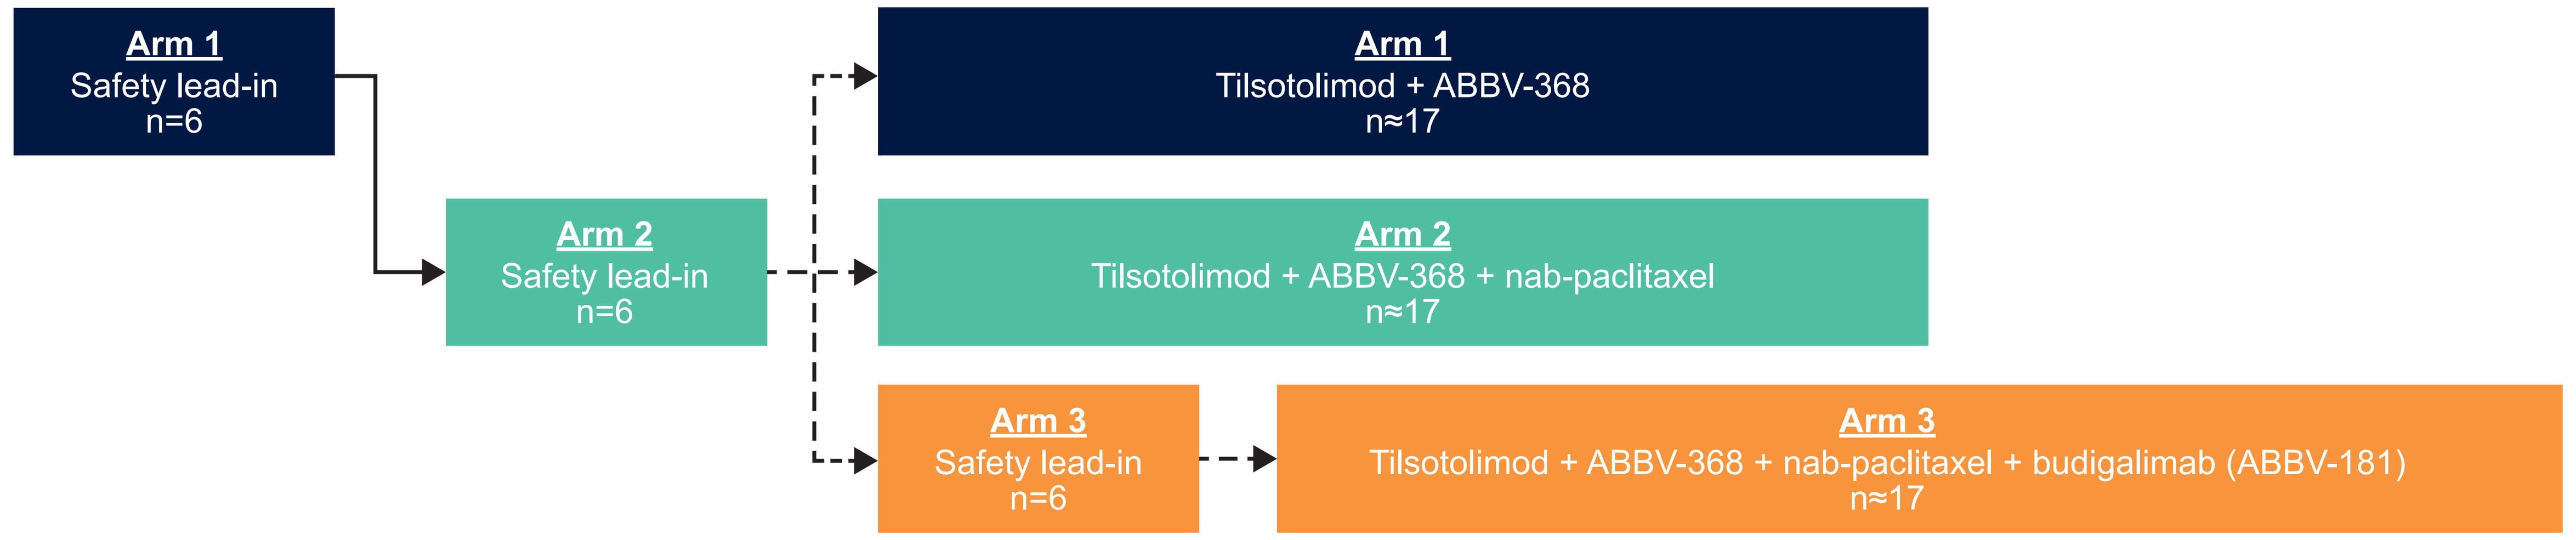
Arm 1
Safety lead-in
n=6
Arm 2
Safety lead-in
n=6
Arm 3
Safety lead-in
n=6
Arm 1
Tilsotolimod + ABBV-368
n≈17
Arm 2
Tilsotolimod + ABBV-368 + nab-paclitaxel
n≈17
Arm 3
Tilsotolimod + ABBV-368 + nab-paclitaxel + budigalimab (ABBV-181)
n≈17

STUDY ASSESSMENT AND STATISTICAL METHODS

- **Safety:** Safety and tolerability will be measured by monitoring adverse events (AEs), vital sign measurements, and clinical laboratory testing (hematology, chemistry, and urinalysis) as per the National Cancer Institute's Common Terminology Criteria for Adverse Events, version 5 (NCI CTCAE, v5)
- **Efficacy**: Objective response rate (ORR), clinical benefit rate (CBR), time to response (TTR), progression-free survival (PFS), and duration of response (DOR) will be assessed according to RECIST v1.1 and iRECIST; tumor assessments will be performed at baseline and multiple timepoints throughout the study until study completion
- **PK:** PK samples to assess serum concentrations of ABBV-368 and budigalimab and plasma concentrations of tilsotolimod will be obtained at multiple timepoints throughout the study
- **Biomarkers**: Biospecimens will be collected at specified timepoints to evaluate receptor occupancy and pharmacodynamic biomarkers in the blood and tissues
- **Immunogenicity**: Antidrug antibody (ADA) assays will be performed and reported as positive or negative

SUMMARY

- This multicenter, international Phase 1b study is investigating the safety, tolerability, and PK of ABBV-368 plus tilsotolimod in combination with other immune-modulating agents in patients with R/M HNSCC
- Preliminary efficacy (ORR, CBR, TTR, PFS, and DOR) based on RECIST 1.1 will be assessed as secondary endpoints
- Preliminary efficacy (ORR, CBR, TTR, PFS, and DOR) based on iRECIST, as well as pharmacodynamics and predictive biomarkers will be assessed as exploratory endpoints

REFERENCES

1. Cancer.gov accessed July 24, 2020 at https://www.cancer.gov/types/head-and-neck/head-neck-fact-sheet#r28.
2. Lala M, et al. *Oral Oncol.* 2018;84:108-120.
3. Sagiv-Barfi I, et al. *Sci Transl Med.* 2018;10(426)eeaan4488.
4. Le Tourneau C, et al. *JITC.* 2019;7(suppl 1, abstr P432):282; DOI: 10.1186/s40425-019-0763-1.
5. Babiker H, et al. AACR. 2020;134.

ACKNOWLEDGMENTS

AbbVie and the authors thank the patients participating in this clinical trial and all study investigators for their contributions.

DISCLOSURES

AbbVie: AbbVie funded this study and participated in the study design, research, analysis, data collection, interpretation of data, reviewing, and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship. Medical writing support was provided by Devon Roll, PhD, of Bio Connections, LLC, and funded by AbbVie.

X. Le: consulting/advisory fees from Eli Lilly, AstraZeneca, EMD-Serono and research funds from Eli Lilly, Boehringer Ingelheim, and Spectrum. **C. Maurice-Dror:** speakers' bureau participation/lecturer for Bristol-Myers Squibb (BMS), MSD, and Medison. **M. Nagasaka:** consultancy/advisory role for AstraZeneca, Daiichi-Sankyo, Takeda, Novartis, and Caris Life Sciences and research funding from Tempus. **M Oliva:** Consulting fees for BMS; investigator for AbbVie-sponsored clinical trials. **M. Patel, D. Da Costa, S. Lambert, Y. Li, M. Blaney,** and **M. McDevitt:** employment with AbbVie and may own AbbVie stock or stock options. **A. Rosenberg:** investigator for AbbVie-sponsored trial and consulting/advisory fees from EMD-Serono. **I. Gluck**, **A. Panwar, K. Gold, T. Dai, J. Grewal, M. Haigentz, C. Le Tourneau, I. Moreno:** investigator for AbbVie-sponsored clinical trials. **M. Gillison:** consulting fees from Bicara, Bayer, Roche, Kura, BioNTech, Shattuck Labs, EMD Serono, Merck, and BMS; research funding from Genocea, Cullinan, BMS, and Kura; and investigator for AbbVie-sponsored clinical trials.

For questions regarding this poster, please contact the corresponding author, Dr Xiuning Le: xle1@mdanderson.org

Copies of this Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ESMO and the author of this poster.

Scan QR code to download an electronic version of this poster presentation (#2195) and other AbbVie ESMO 2020 scientific presentations.
QR code expiration: September 17, 2021.